Apabetalone, a BET Bromodomain Inhibitor, Suppresses Inflammatory Mediators in Microglia and Vascular Endothelial Cells that Contribute to Neurodegenerative Disease

Ewelina Kulikowski[1], Emily Daze[1], Sylwia Wasiak[1], Dean Gilham[1], Laura M. Tsujikawa[1], Li Fu[1], Brooke D. Rakai[1], Stephanie C. Stotz[1], Christopher Halliday[1], Ravi Jahagirdar[1], Norman C. W. Wong[1], Michael Sweeney[2] and Jan O. Johansson[2]

[1]Resverlogix Corp. Calgary, Canada and [2]Resverlogix Inc. San Francisco, USA.

Background

Apabetalone is an orally available small molecule in phase 3 trials for cardiovascular disease (CVD). As an inhibitor of bromodomain and extraterminal domain (BET) proteins, apabetalone regulates gene expression through an epigenetic mechanism. Clinical trials in CVD patients and preclinical models demonstrate peripheral anti-inflammatory effects of apabetalone treatment. Systemic or localized peripheral inflammatory injury can lead to transmission of neural signals as well as infiltration of peripheral molecules and immune cells into the central nervous system (CNS). This contributes to microglial pro-inflammatory activation and neurodegeneration.

Objective

We evaluated the therapeutic potential of apabetalone in pre-clinical models of neuroinflammation. We assessed responses to apabetalone in human brain endothelial cells, microglial cells and brains of endotoxemic mice.

Results

Vascular endothelial cells convey inflammatory responses from the periphery to the CNS while activated microglial cells propagate inflammation in the CNS, ultimately leading to neuronal injury. Treatment of the human brain endothelial cell line hCMEC/D3 and primary endothelial cells HBMVECs with TNF-α and interferon-γ showed an upregulation of markers of inflammation and vascular activation such as interleukin-6, interleukin-1β, monocyte chemoattractant protein 1 (MCP-1), VCAM-1 and E-selectin. Apabetalone dose dependently opposed this induction at the gene expression level. The surface adhesion proteins VCAM-1 and E-selectin were also reduced. Treatment of BV-2 cells *in vitro* showed that apabetalone dose dependently opposed lipopolysaccharide (LPS) and interferon-γ mediated induction of key contributors to neurodegenerative processes such as IL-6, IL-1β, complement C3 and C1q. Peripheral LPS injection in 8-week C57BL6 male mice (10 ug/animal) elicits an inflammatory response in the CNS after 24h. Pre-treatment with apabetalone (7 days, 150 mg/kg, b.i.d.) countered the upregulation of endothelial adhesion molecules E-selectin and ICAM-1, the monocyte marker CCR2 and the macrophage marker CD68 in the mouse brain.

Mechanism of Action

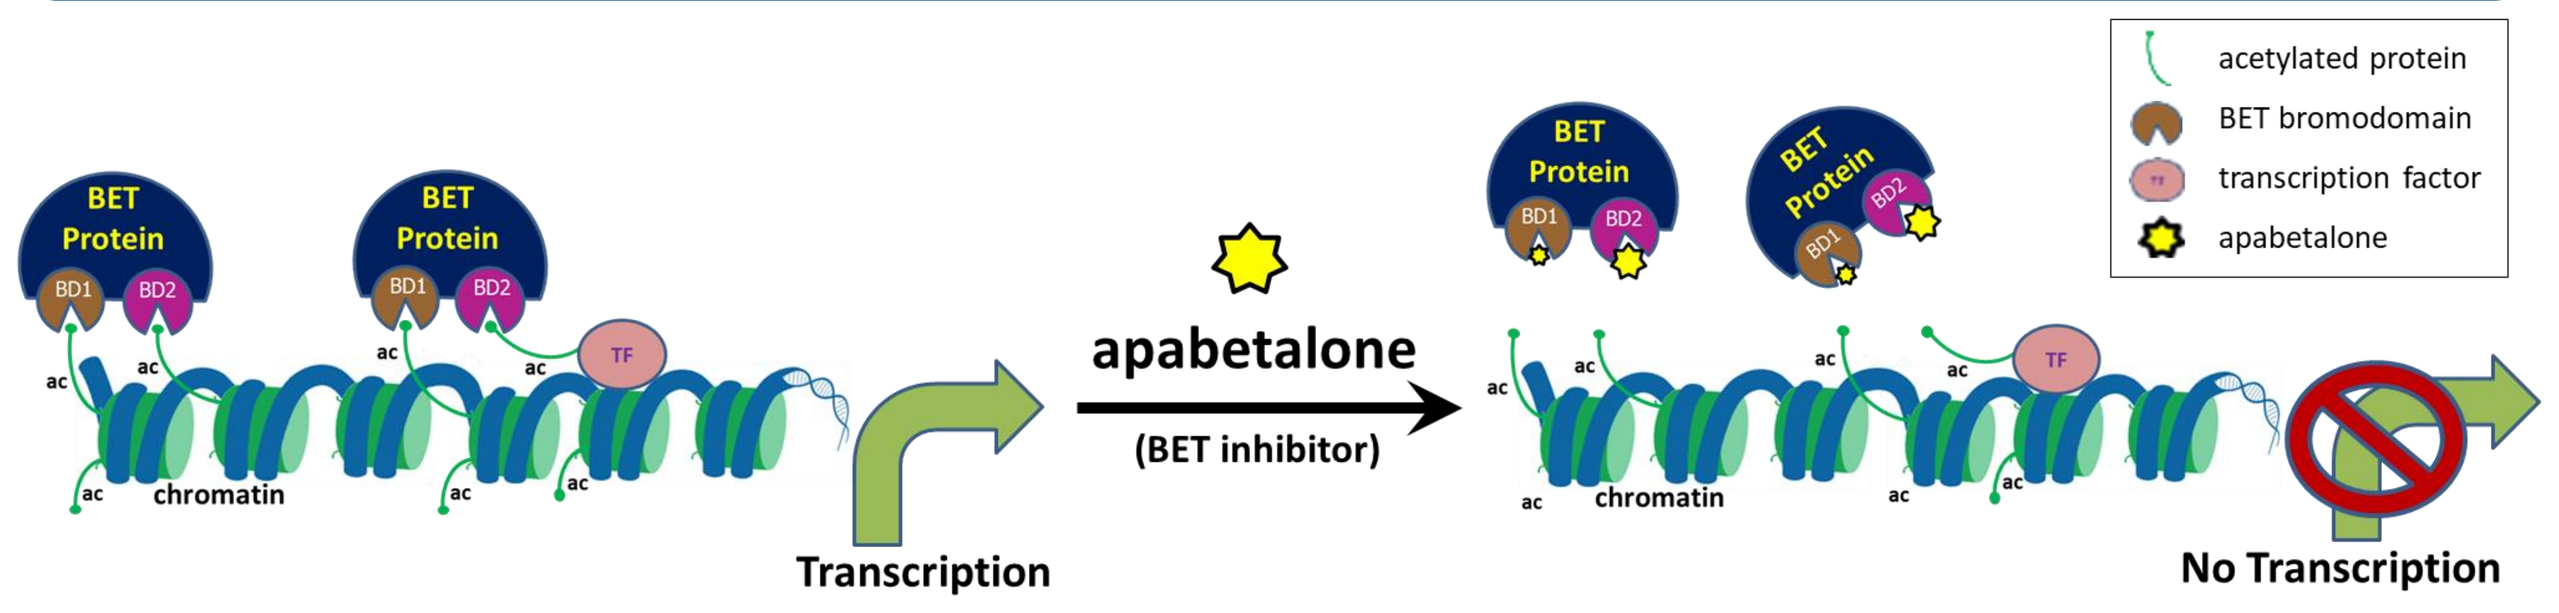
BET Protein
BD1
BD2
TF
ac
chromatin
Transcription
apabetalone
(BET inhibitor)
No Transcription
acetylated protein
BET bromodomain
transcription factor
apabetalone

BET proteins, such as BRD4, bind acetylated lysine (ac) on histones or transcription factors (TF) via bromodomains (BD), and recruit transcriptional machinery to drive expression of BET sensitive genes. Apabetalone targets bromodomains in BET proteins, causing release from chromatin and downregulation of BET sensitive gene expression.

Yellow star size indicates selectivity of apabetalone for bromodomain 2.

Apabetalone Suppresses Expression of Inflammatory Mediators in Stimulated Brain Endothelial Cells

Endothelial cells were stimulated with 10 ng/mL TNFα and IFNγ in the presence or absence of BET inhibitors for 4h or 24h.
Gene expression was analyzed by real-time PCR.

hCMEC/C3 (4h) | HBMVECs (24h)

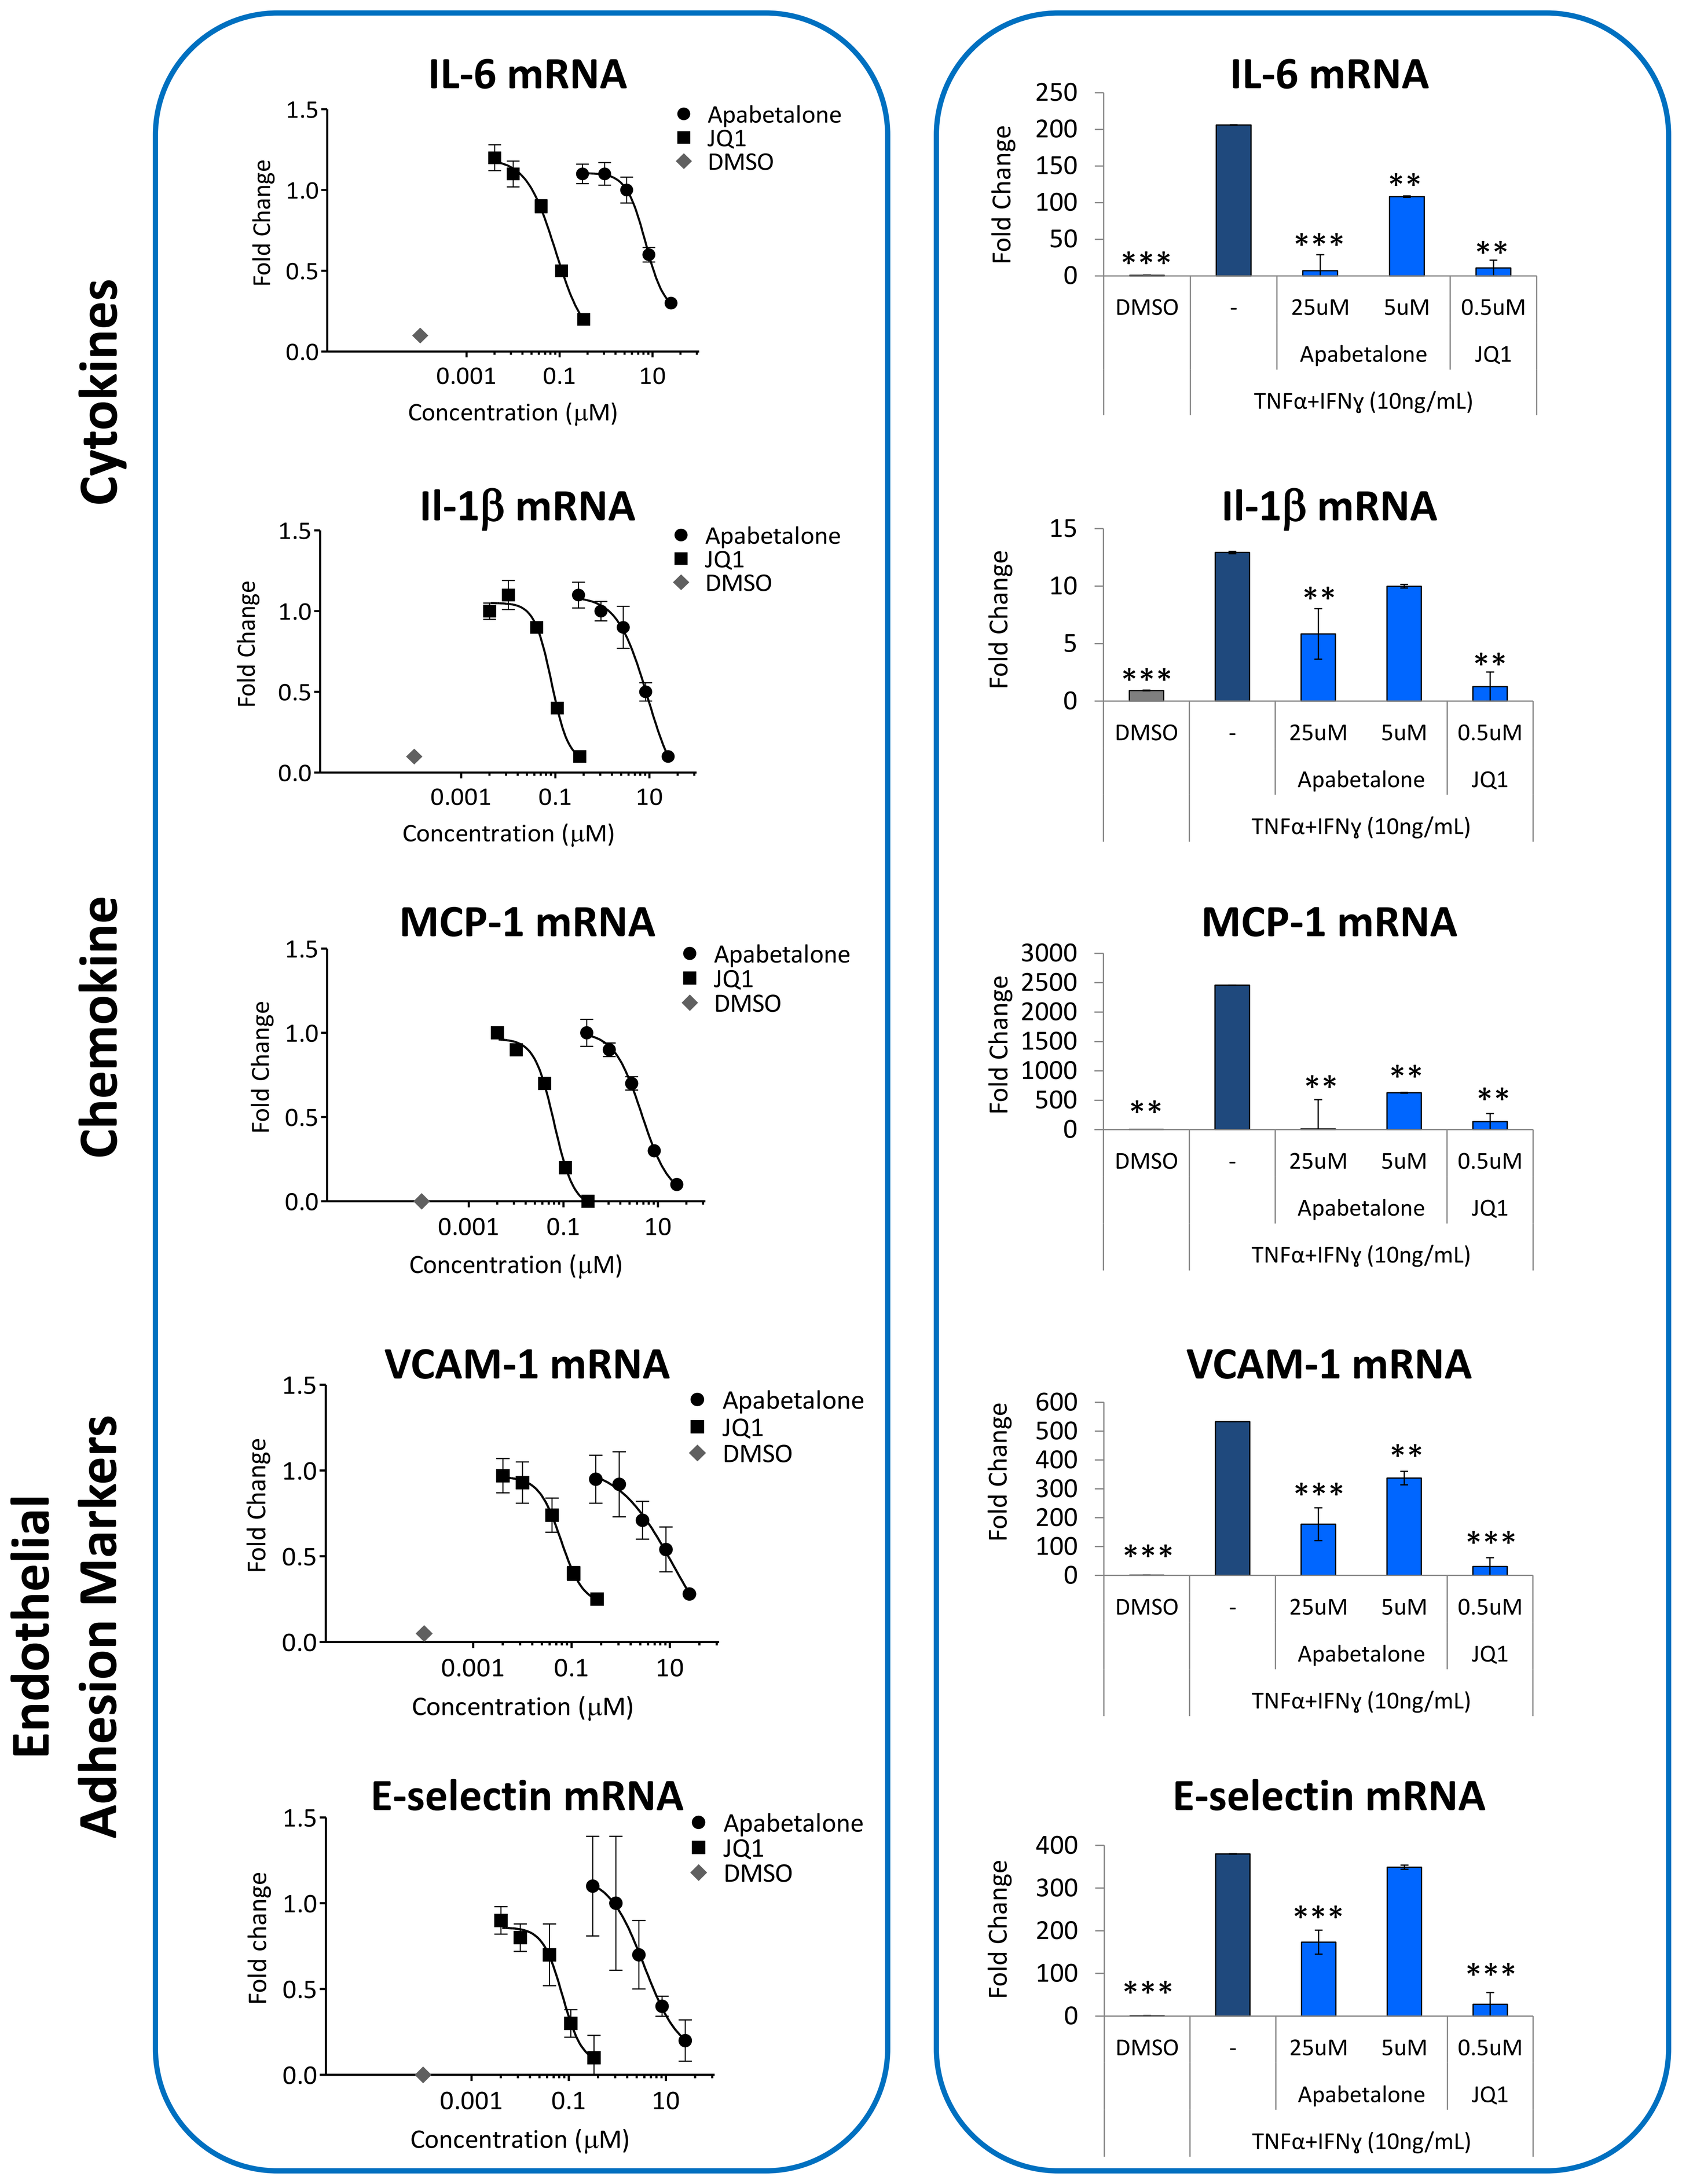
Cytokines
Chemokine
Endothelial Adhesion Markers
IL-6 mRNA
Il-1β mRNA
MCP-1 mRNA
VCAM-1 mRNA
E-selectin mRNA
Apabetalone
JQ1
DMSO
Fold Change
Concentration (μM)
DMSO
-
25uM
5uM
0.5uM
Apabetalone
JQ1
TNFα+IFNγ (10ng/mL)

HBMVECs were stimulated with 10 ng/mL TNFα and IFNγ (4h) ± apabetalone. Protein surface expression was measured by FACS.

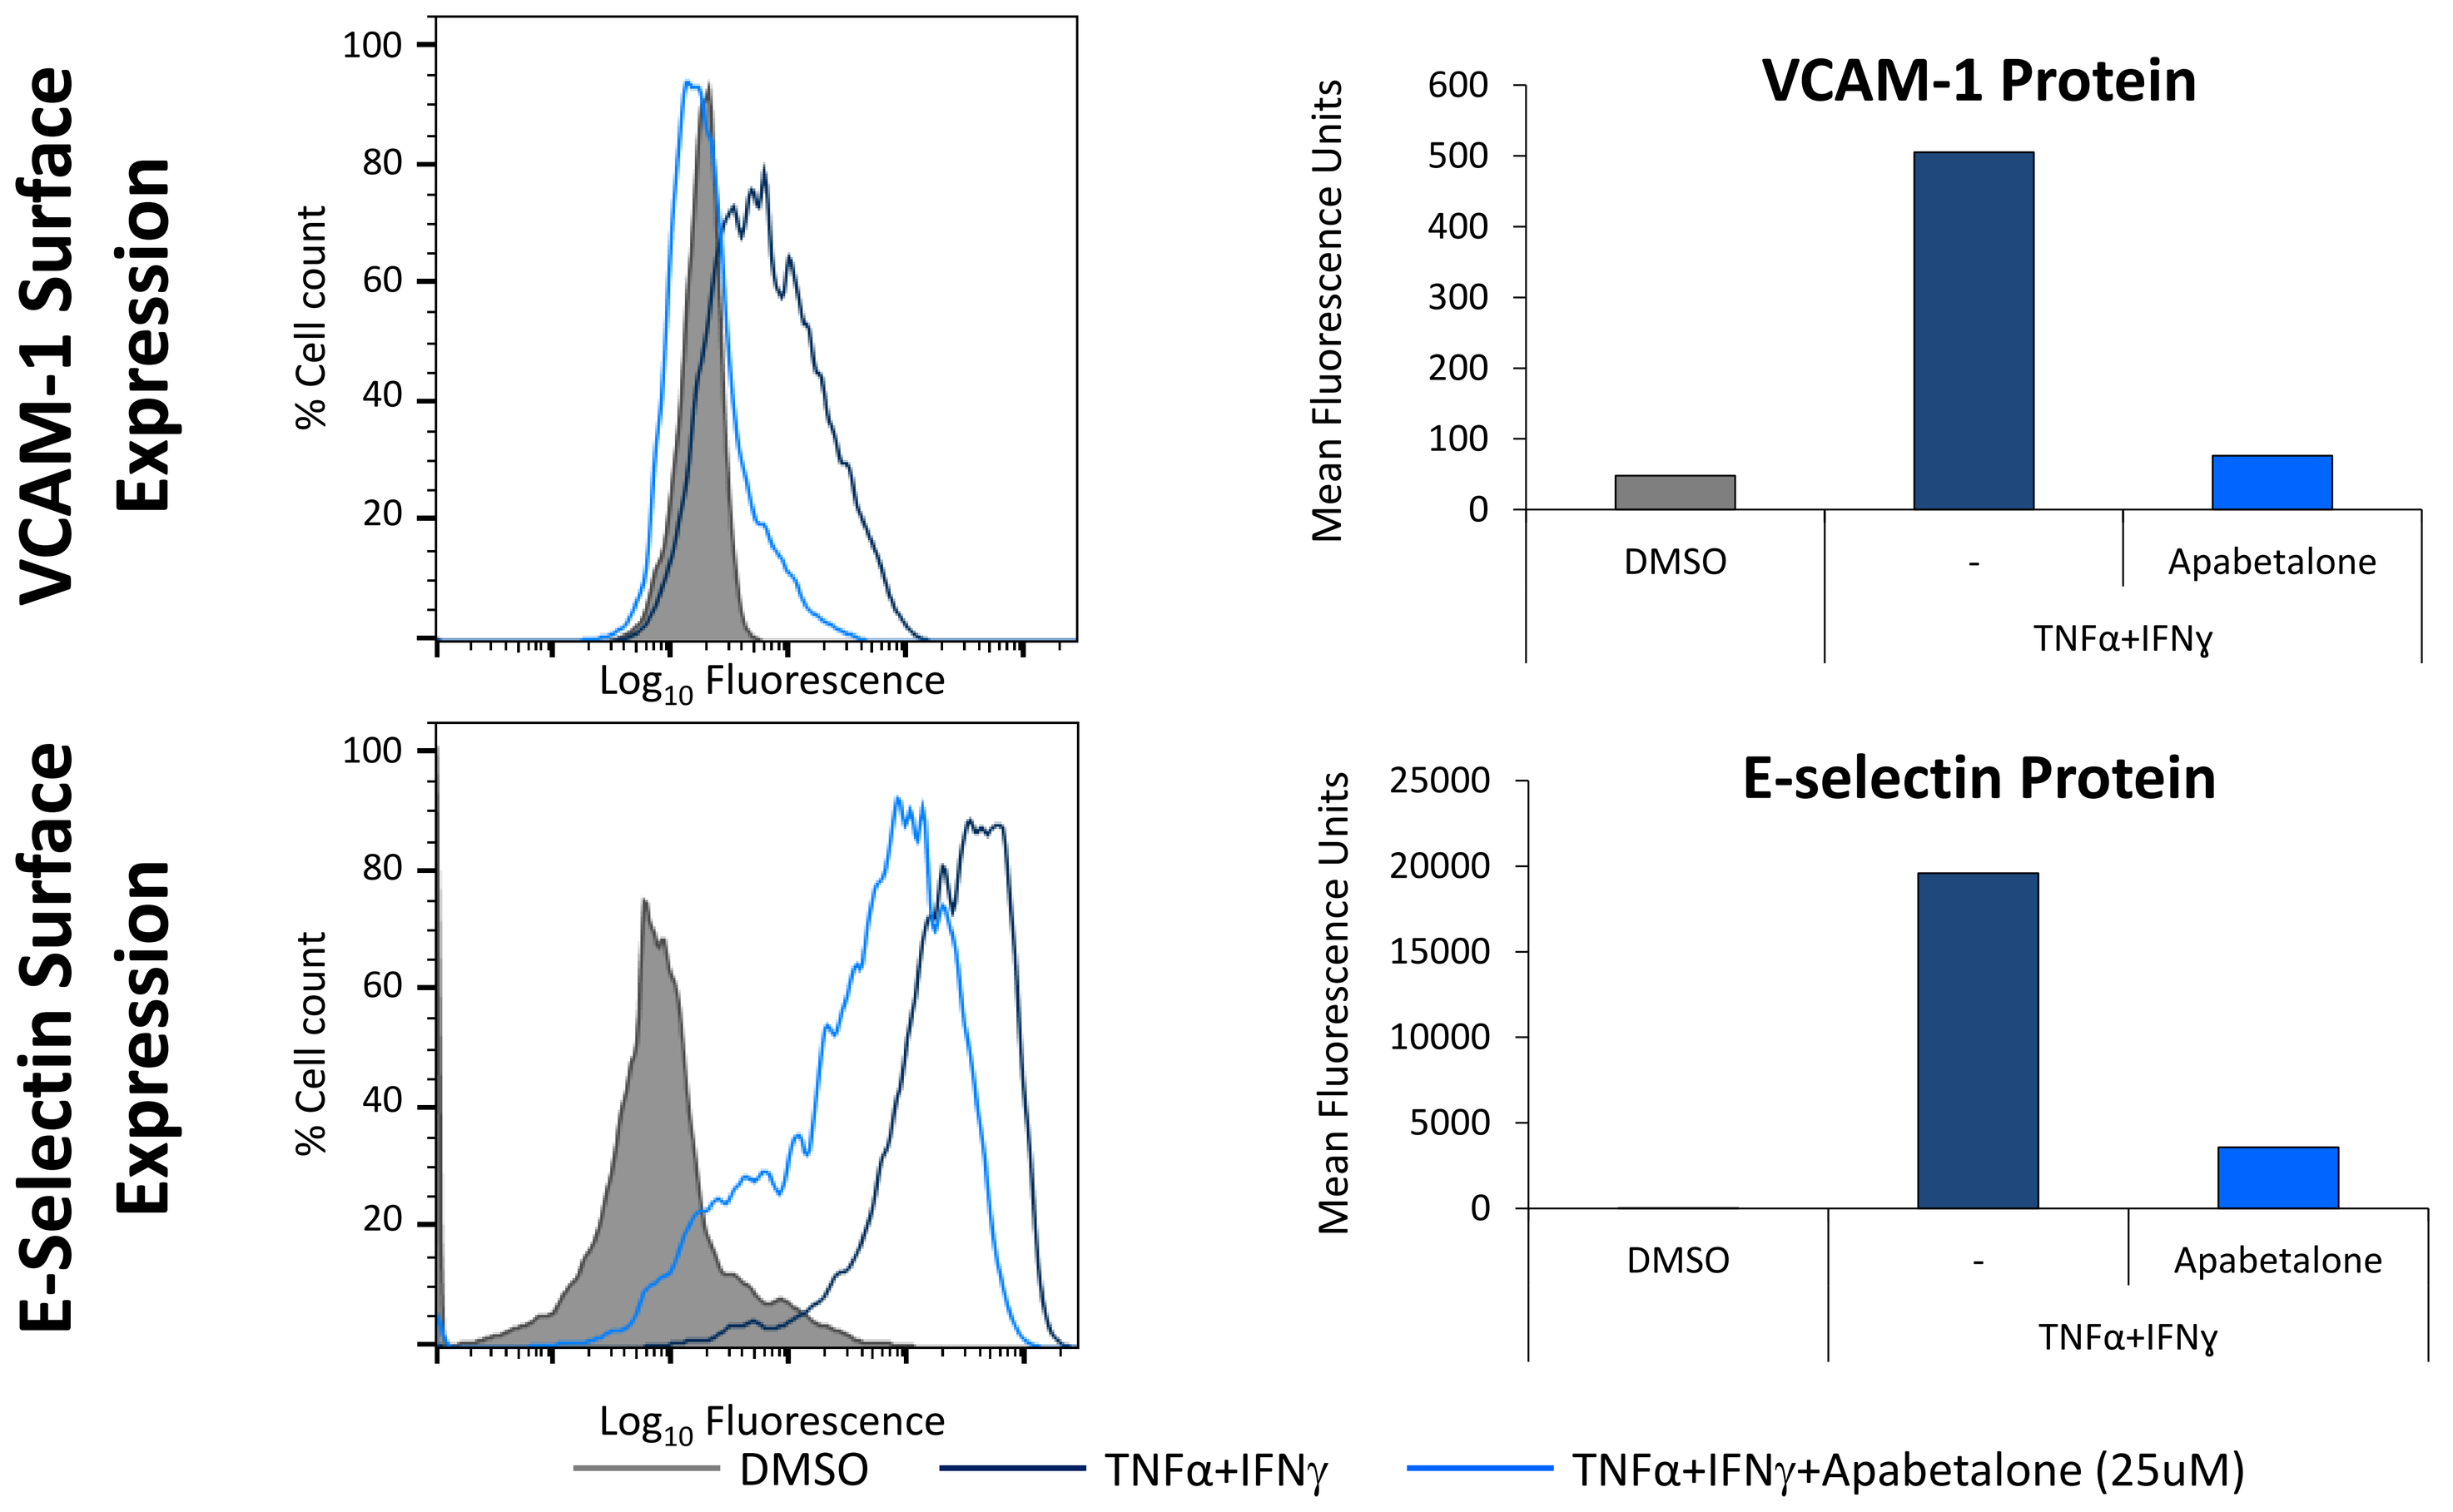
VCAM-1 Surface Expression
E-Selectin Surface Expression
% Cell count
Log10 Fluorescence
VCAM-1 Protein
E-selectin Protein
Mean Fluorescence Units
DMSO
-
Apabetalone
TNFα+IFNγ
DMSO
TNFα+IFNγ
TNFα+IFNγ+Apabetalone (25uM)

Statistical significance determined with a Student's *t*-test * $p<0.05$; ** $p<0.01$; *** $p<0.001$

Apabetalone Suppresses Expression of Inflammatory Mediators in Stimulated BV-2 Microglial Cells

BV-2 cells were stimulated with 100 ng/mL LPS & 5 ng/mL IFNγ in the presence or absence of BET inhibitors for 2h.
Gene expression was analyzed by real-time PCR.

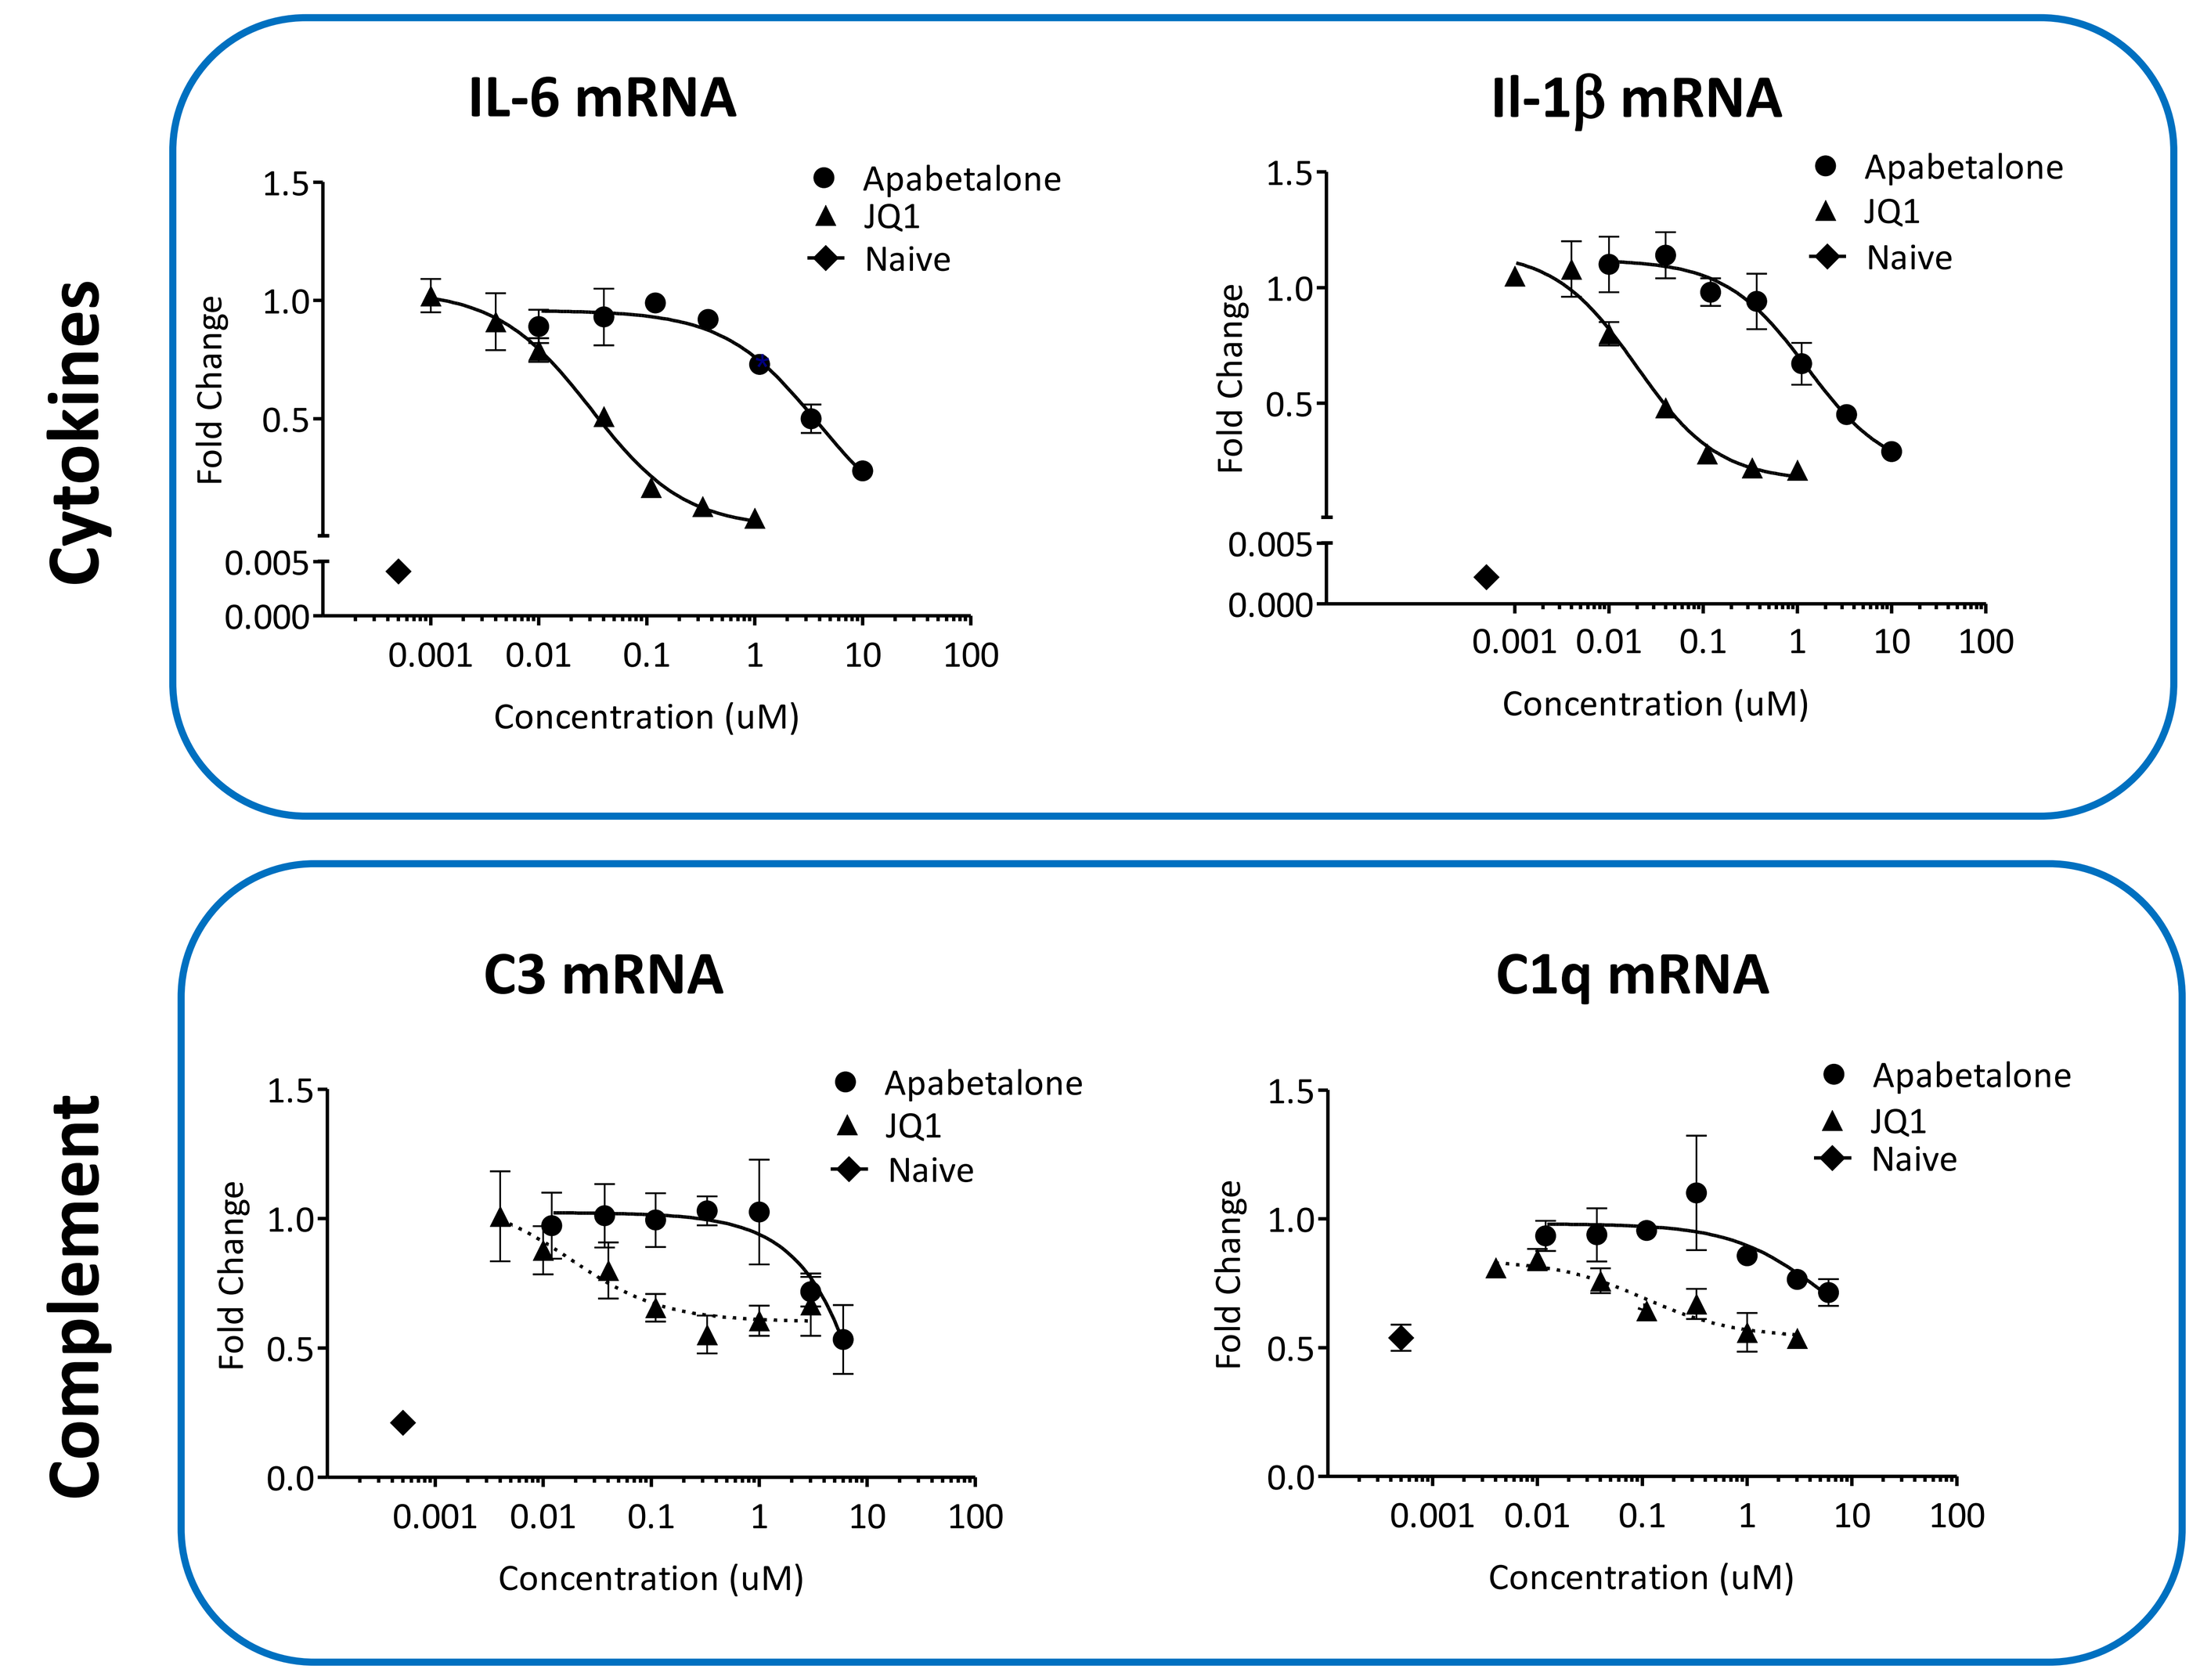
Cytokines
Complement
IL-6 mRNA
Il-1β mRNA
C3 mRNA
C1q mRNA
Apabetalone
JQ1
Naive
Fold Change
Concentration (uM)

Note: Naïve cells did not receive cytokine or BETi treatment

Apabetalone Reduces Expression of Inflammation Markers in the Brain of Endotoxemic Mice (24h Post LPS Stimulation)

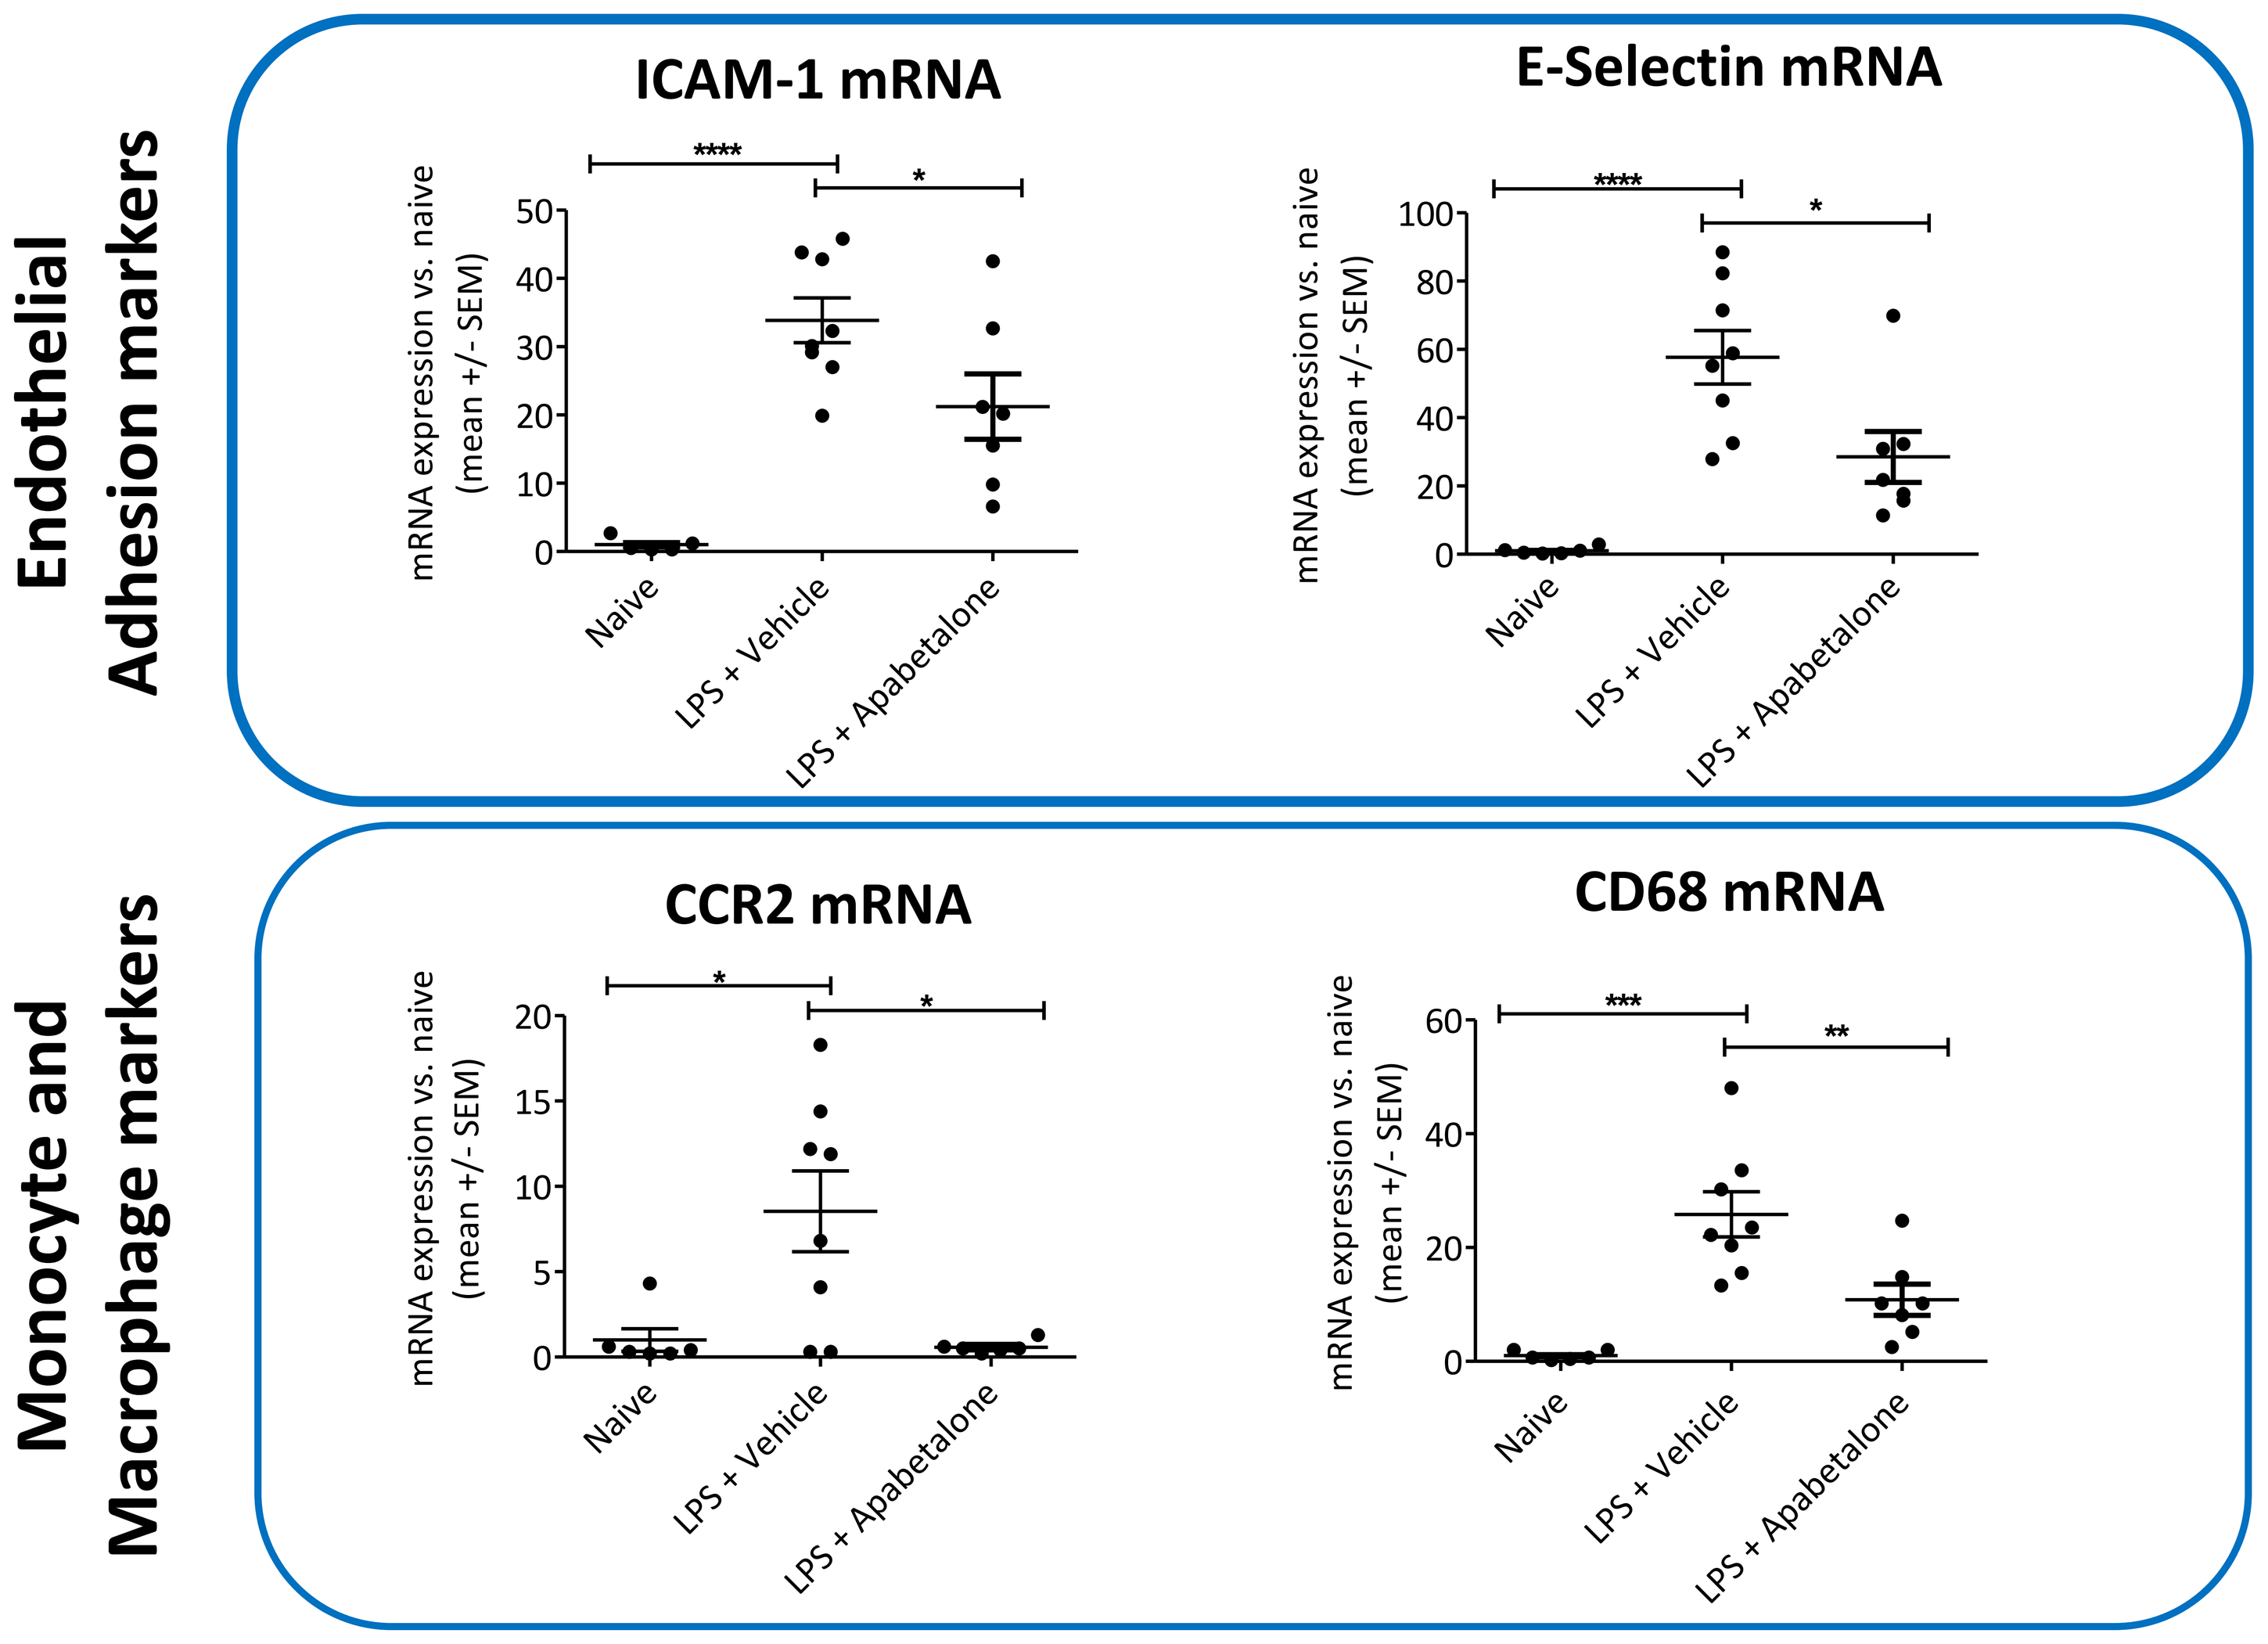
Endothelial Adhesion markers
Monocyte and Macrophage markers
ICAM-1 mRNA
E-Selectin mRNA
CCR2 mRNA
CD68 mRNA
mRNA expression vs. naive (mean +/- SEM)
Naive
LPS + Vehicle
LPS + Apabetalone

Note: Naïve animals did not receive LPS or BETi treatment

Conclusions

- **In CNS cell models and inflamed mice, the epigenetic inhibitor apabetalone can counter inflammatory expression of cytokines, chemokines and markers of endothelial and microglial activation associated with neuroinflammation and cognitive dysfunction.**
- **The effect of apabetalone on cognition is currently being evaluated with MoCA in participants ≥70 years of age enrolled in the phase 3 BETonMACE trial focusing on cardiovascular outcomes in patients with CVD and diabetes.**