The Epigenetic Inhibitor Apabetalone Downregulates Brain Endothelial and Microglial Cell Activation that Contributes To Neurodegenerative Disease

Ewelina Kulikowski[1], Emily Daze[1], Sylwia Wasiak[1], Li Fu[1], Dean Gilham[1], Laura M. Tsujikawa[1], Brooke D. Rakai[1], Stephanie C. Stotz[1], Christopher D. Sarsons[1], Deborah Struder[3], Kristina Rinker[3], Ravi Jahagirdar[1], Norman C. W. Wong[1], Michael Sweeney[2] and Jan O. Johansson[2]

[1]Resverlogix Corp. Calgary, AB Canada, [2]Resverlogix Inc. San Francisco, CA USA, [3]Department of Chemical and Petroleum Engineering, Calgary, AB Canada

Background

Systemic or localized peripheral inflammatory injury can lead to transmission of neural signals as well as infiltration of peripheral molecules and immune cells into the central nervous system (CNS). This contributes to microglial pro-inflammatory activation and neurodegeneration. Apabetalone is an orally available small molecule in phase 3 trials for cardiovascular disease (CVD). As an inhibitor of bromodomain and extraterminal domain (BET) proteins, apabetalone regulates gene expression through an epigenetic mechanism. Clinical trials in CVD patients and preclinical models demonstrate peripheral anti-inflammatory effects of apabetalone treatment.

Objective

We evaluated the therapeutic potential of apabetalone in pre-clinical models of neuroinflammation.

Methods

Human brain microvascular endothelial cells (HBMVECs) were stimulated with cytokines TNFα and IFNγ ± apabetalone or JQ1 (BETi) and assayed for gene expression, surface protein levels, and THP-1 monocyte adhesion under flow. BV-2 microglia responses to lipopolysaccharide (LPS) and IFNγ ± BETi were examined. C57BL/6 mice treated with 150 mg/kg apabetalone for 7 days received 10 µg LPS intraperitoneally, followed by brain mRNA analysis on day 8.

Results

In cytokine stimulated HBMVECs, apabetalone dose dependently reduced the induction of vascular activation marker mRNAs including IL-6, IL-1β, MCP-1, VCAM1 and E-selectin. Surface expression of VCAM1 and E-selectin was also reduced, leading to decreased adhesion of HBMVECs with THP-1 monocytes under laminar flow. In BV-2 microglial cells, apabetalone opposed LPS and IFNγ mediated induction of IL-6, IL-1β, complement C3 and C1q. Peripheral LPS injection in mice provokes inflammatory responses in the brain, where apabetalone attenuated the LPS-induced mRNA expression of endothelial, monocyte and macrophage adhesion molecules E-selectin, ICAM, CCR2, and CD68.

Mechanism of Action

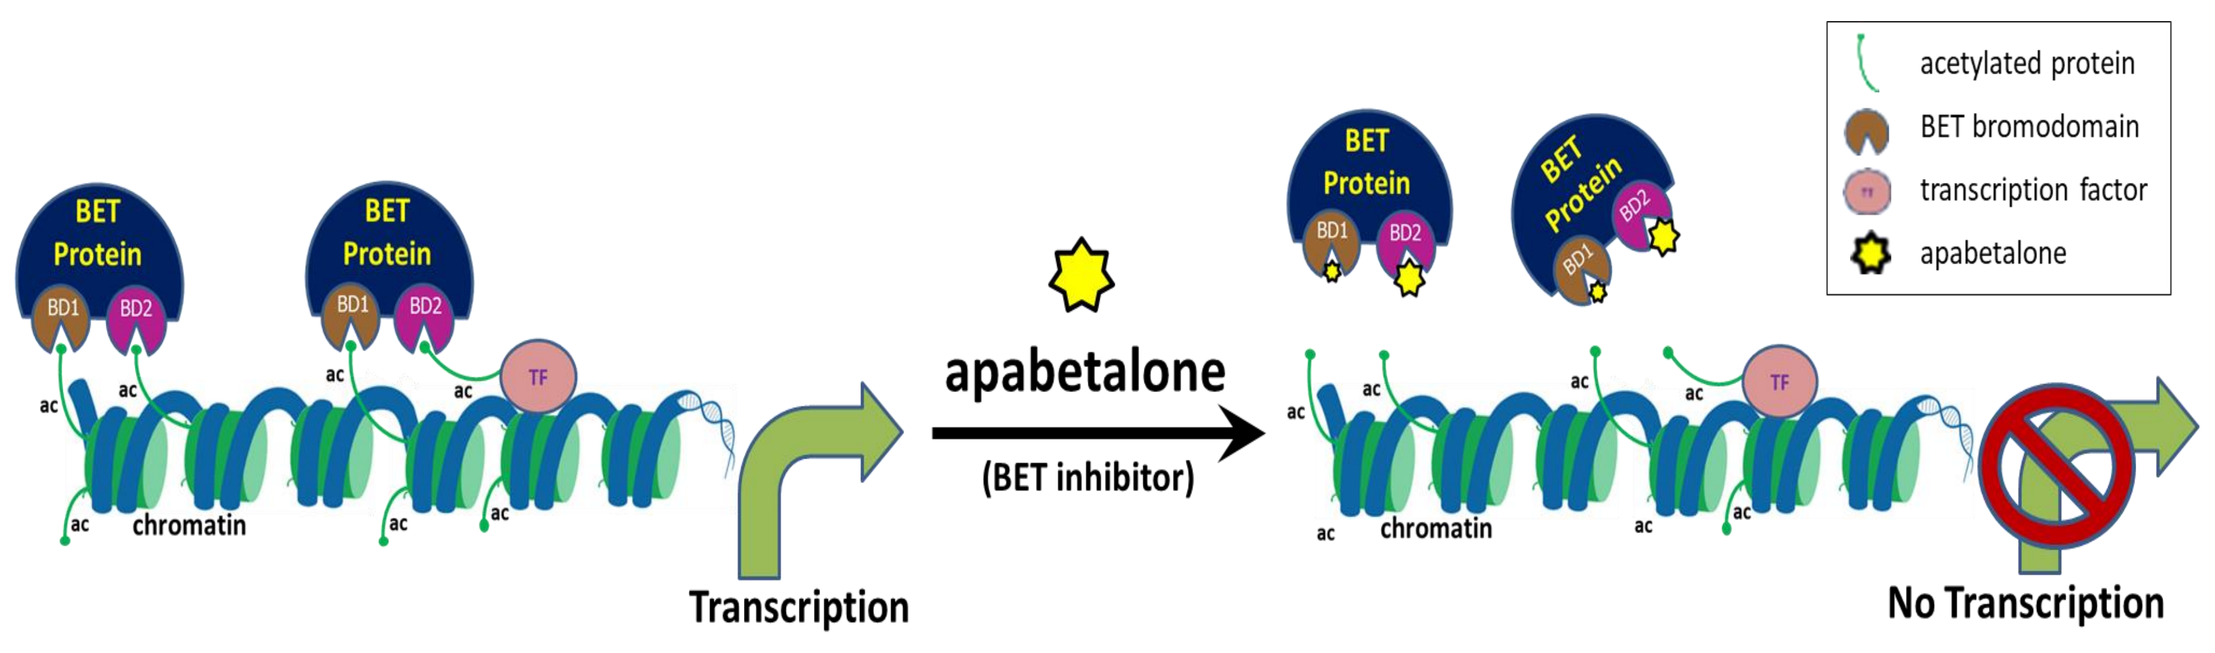
acetylated protein
BET bromodomain
transcription factor
apabetalone
BET Protein
BD1
BD2
TF
ac
chromatin
Transcription
apabetalone
(BET inhibitor)
No Transcription

BET proteins, such as BRD4, bind acetylated lysine (ac) on histones or transcription factors (TF) via bromodomains (BD), and recruit transcriptional machinery to drive expression of BET sensitive genes. Apabetalone targets bromodomains in BET proteins, causing release from chromatin and downregulation of BET sensitive gene expression.

Yellow star size indicates selectivity of apabetalone for bromodomain 2 (BD2).

Leukocyte Infiltration into the Central Nervous System Contributes to Neuroinflammation

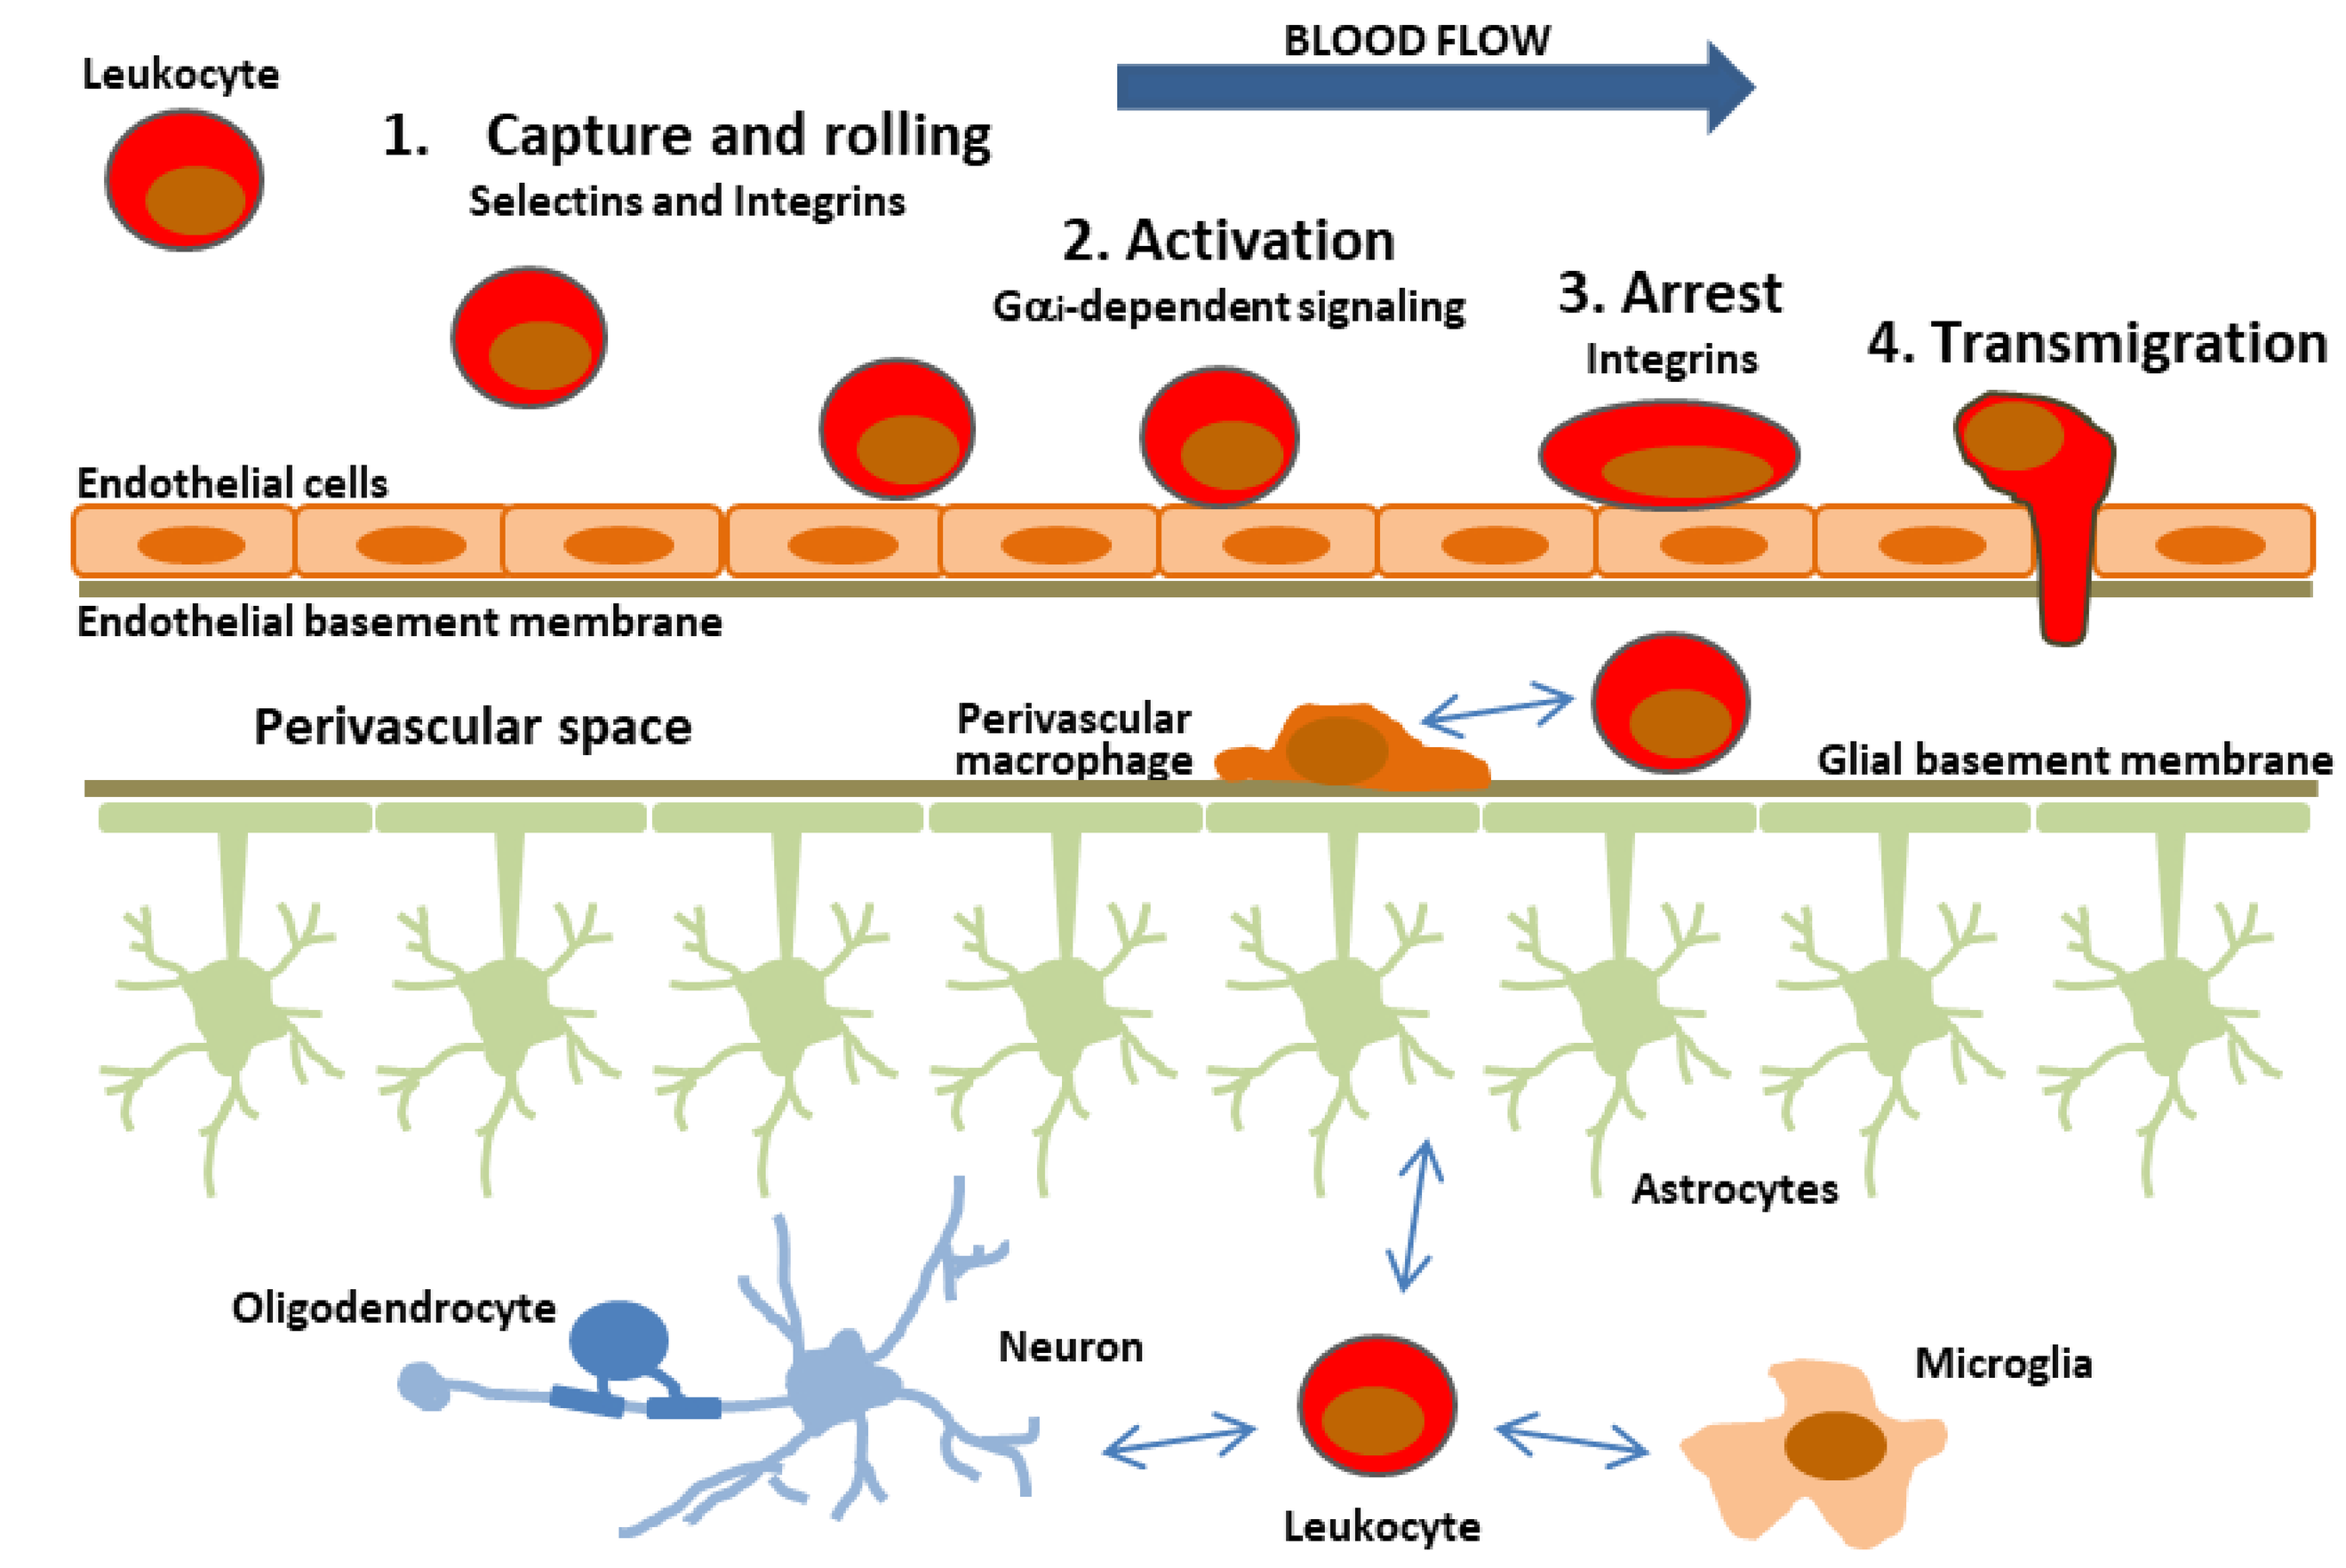
Leukocyte
BLOOD FLOW
1. Capture and rolling
Selectins and Integrins
2. Activation
Gαi-dependent signaling
3. Arrest
Integrins
4. Transmigration
Endothelial cells
Endothelial basement membrane
Perivascular space
Perivascular macrophage
Glial basement membrane
Astrocytes
Oligodendrocyte
Neuron
Leukocyte
Microglia

Adapted from Rossi et al., 2011, Journal of Leukocyte Biology

Apabetalone Suppresses Expression of Inflammatory Mediators in Brain Endothelial Cells

Primary human brain microvascular endothelial cells (HBMVECs) were stimulated for 24h with TNFα and IFNγ ± BETi. Cytokine gene expression was analyzed by real-time PCR.

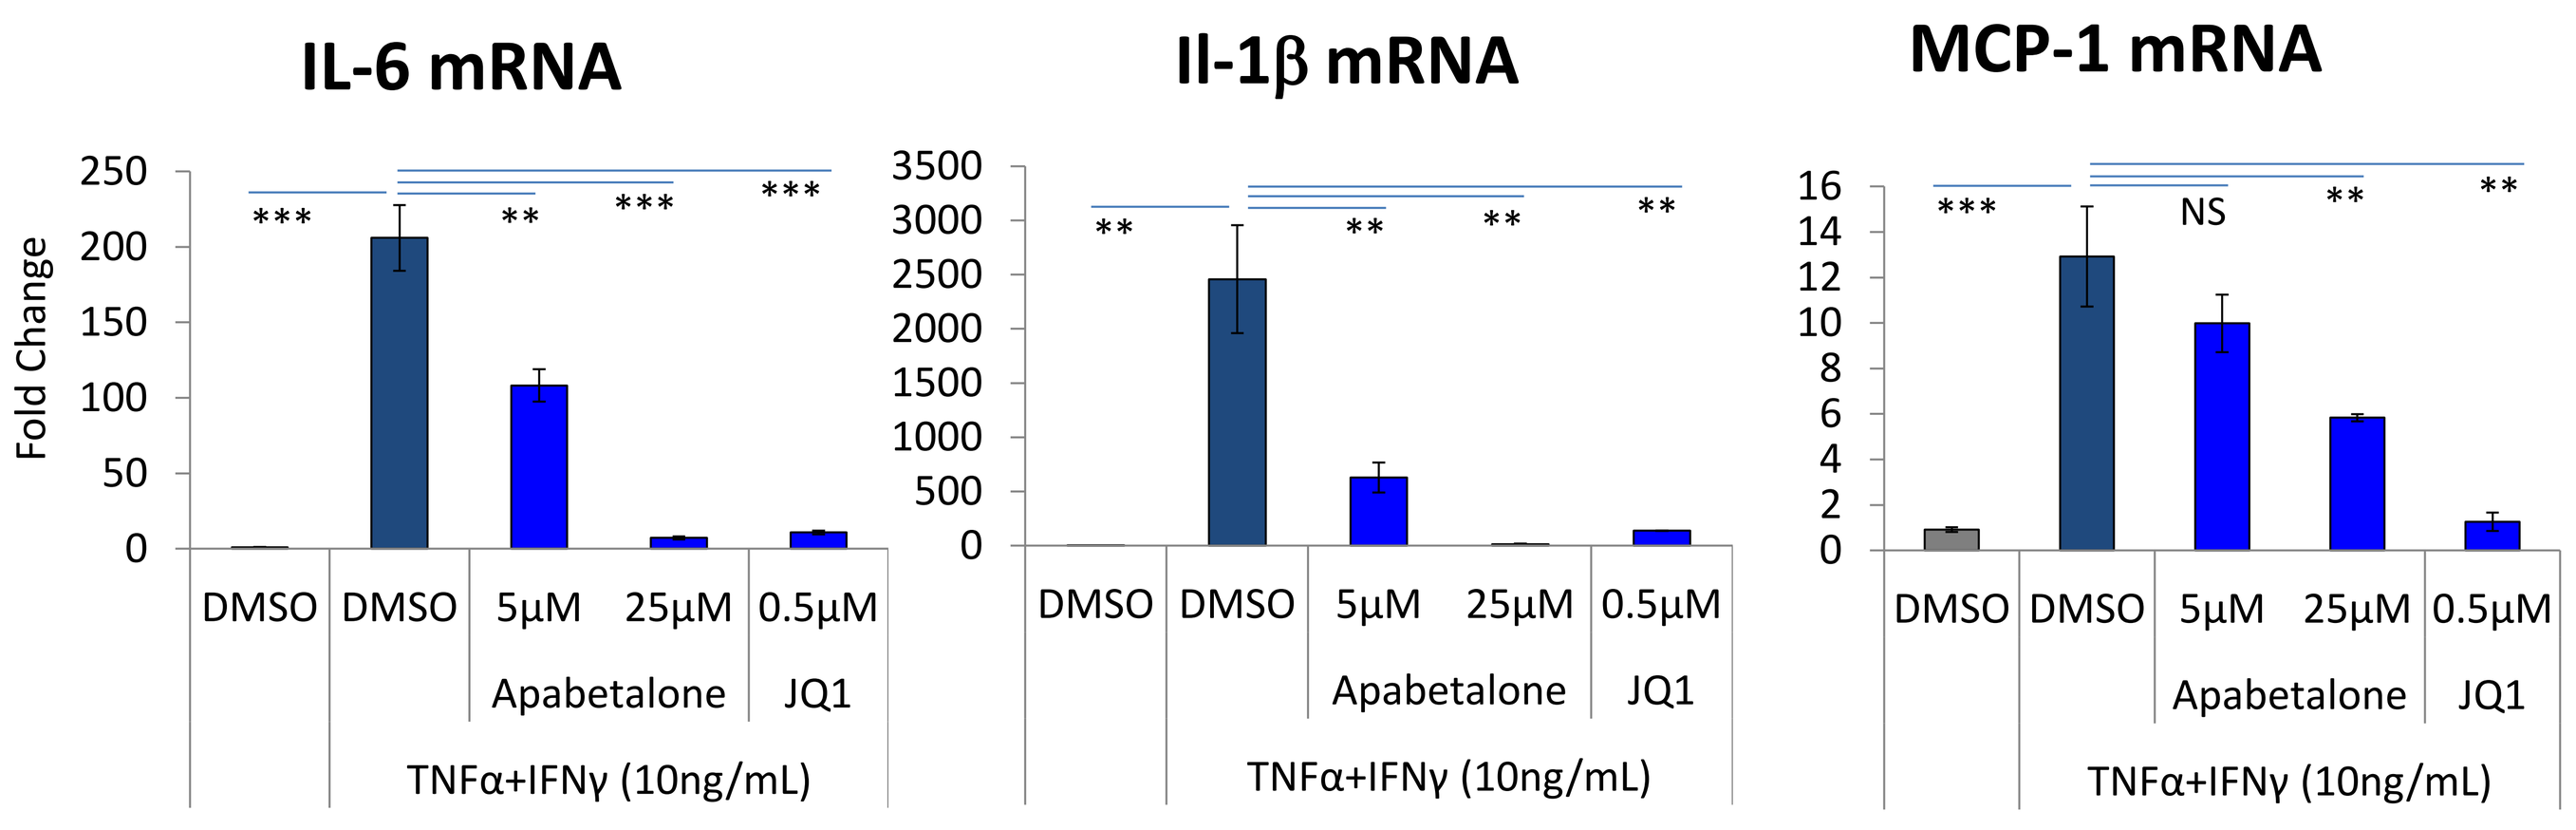
IL-6 mRNA
Il-1β mRNA
MCP-1 mRNA
Fold Change
DMSO
5µM
25µM
0.5µM
Apabetalone
JQ1
TNFα+IFNγ (10ng/mL)
NS

Statistical significance determined with a Student's *t*-test * p<0.05; **p<0.01; ***p<0.001; NS-non significant

HBMVECs were stimulated with TNFα and IFNγ for 24h ± BETi. Adhesion protein surface expression was measured by FACS.

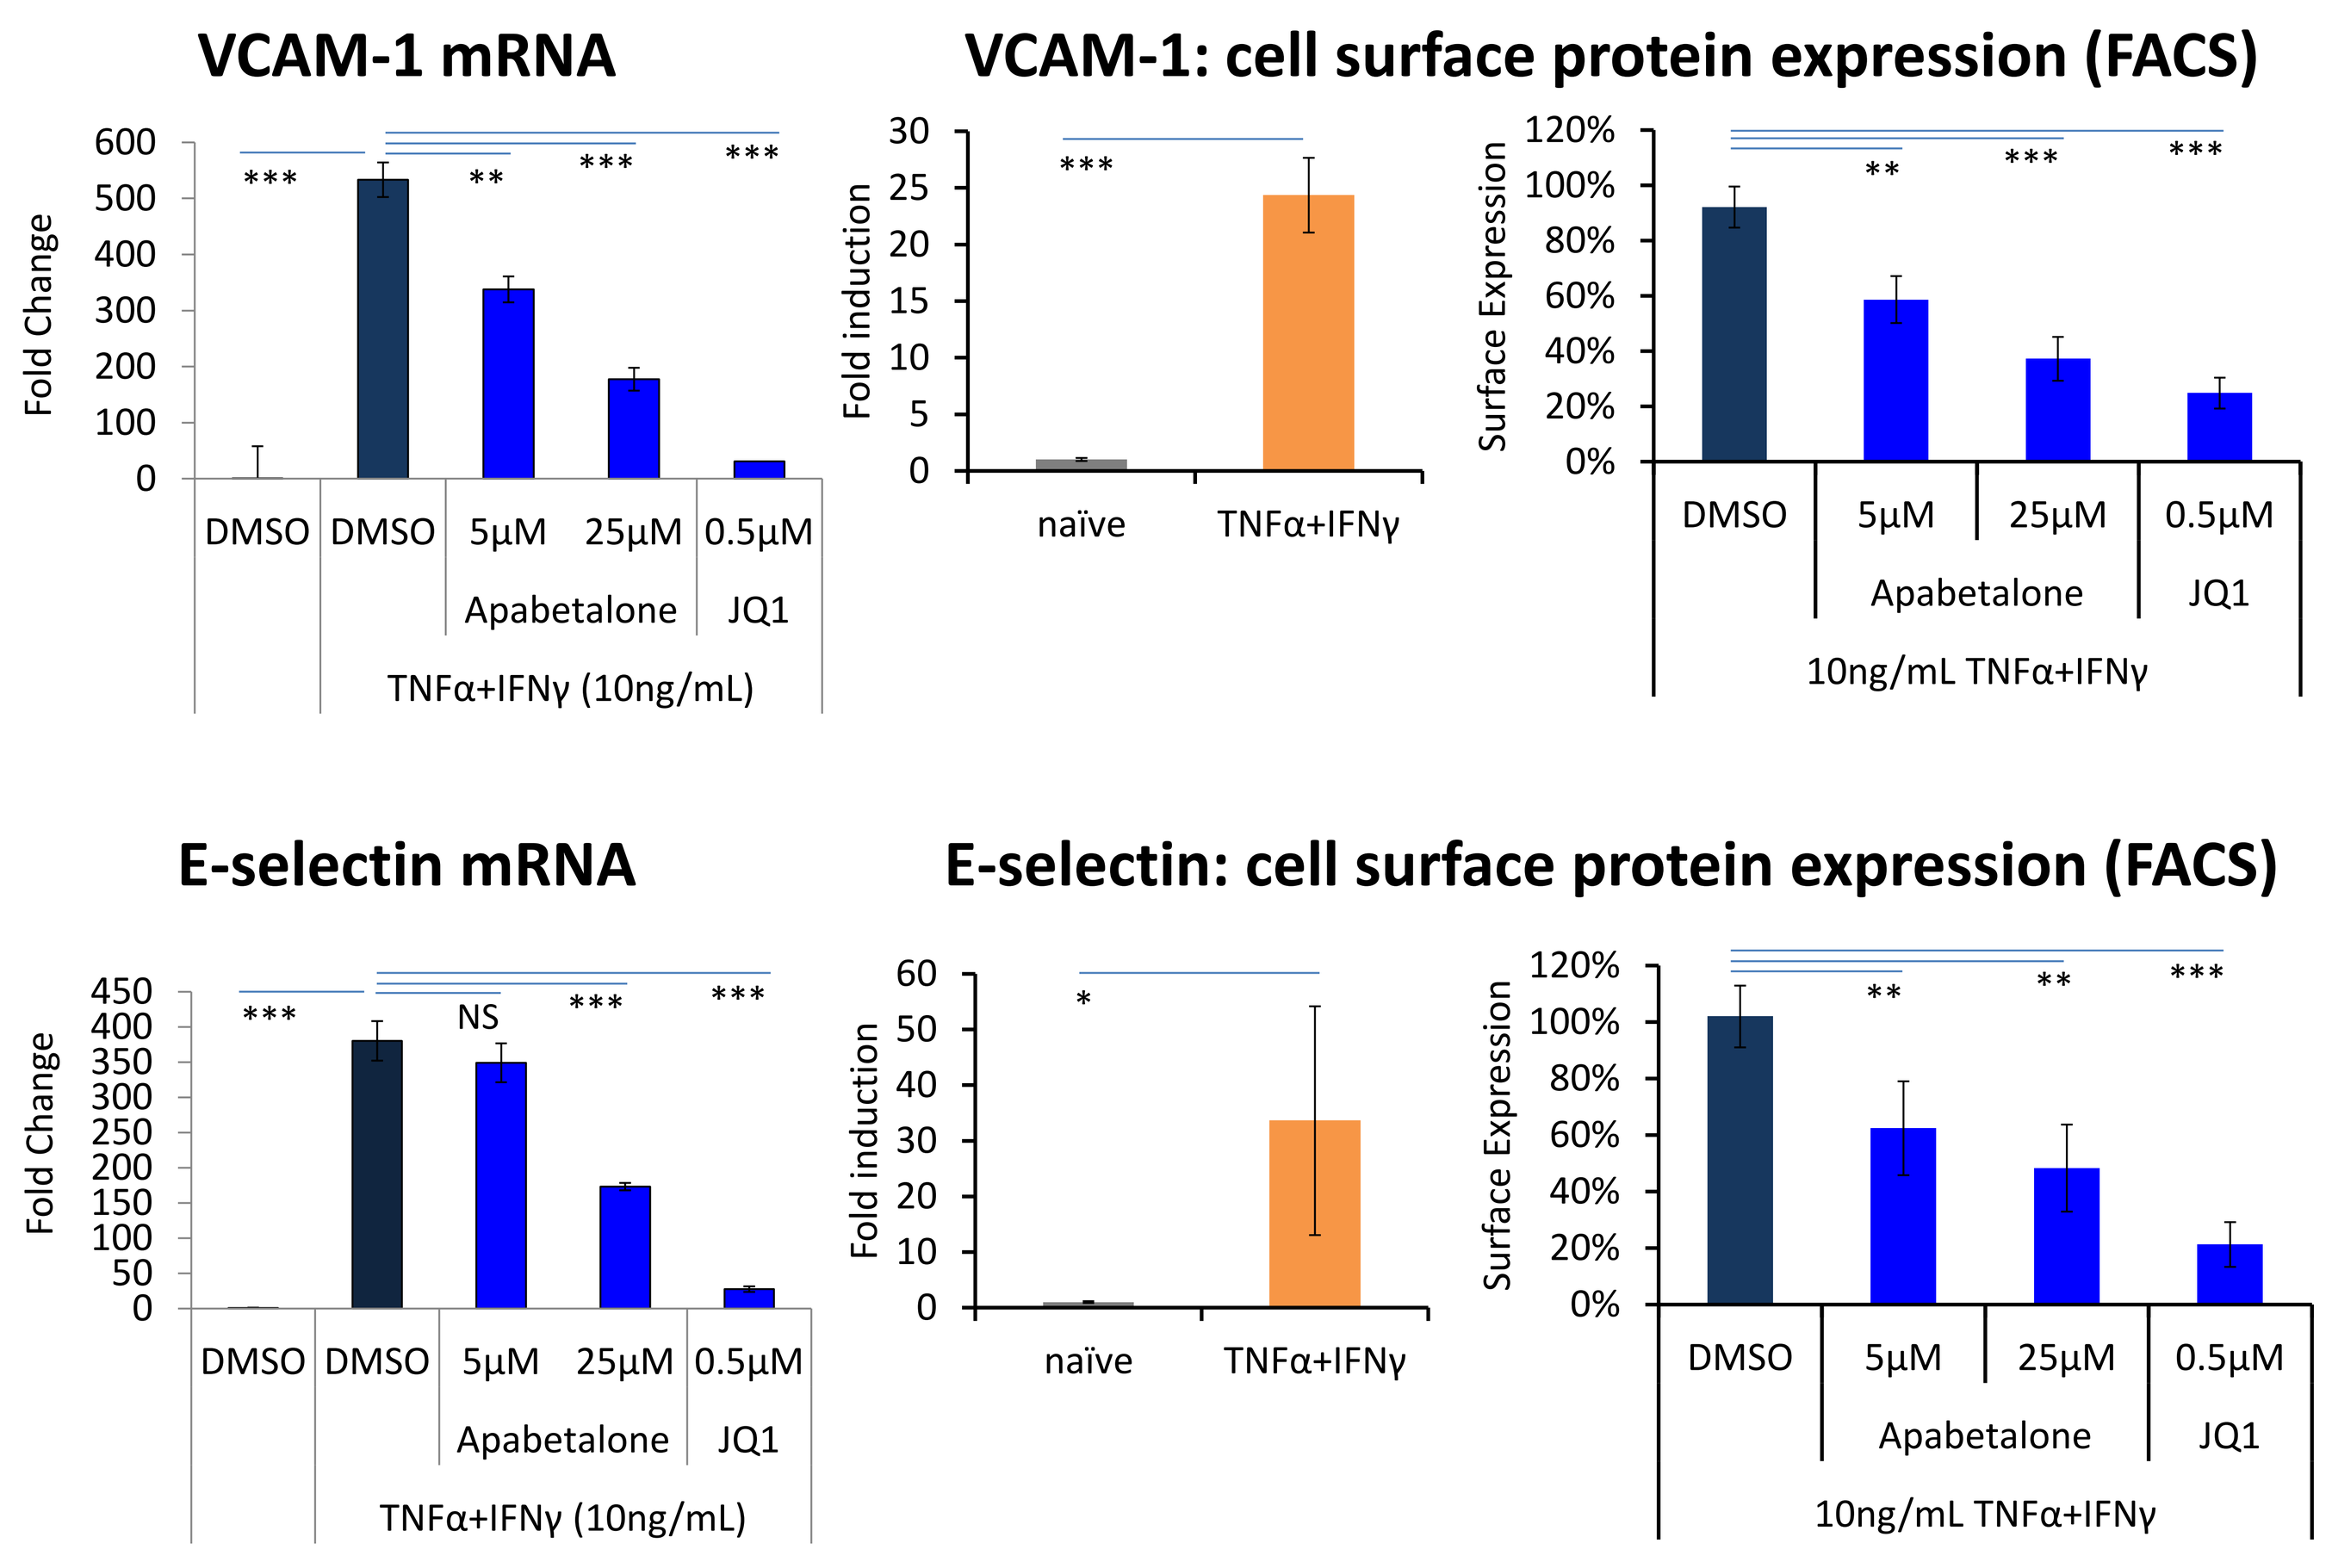
VCAM-1 mRNA
VCAM-1: cell surface protein expression (FACS)
E-selectin mRNA
E-selectin: cell surface protein expression (FACS)
Fold Change
Fold induction
Surface Expression
DMSO
5µM
25µM
0.5µM
Apabetalone
JQ1
TNFα+IFNγ (10ng/mL)
naïve
TNFα+IFNγ
10ng/mL TNFα+IFNγ
NS

Statistical significance determined with a Student's *t*-test * p<0.05; **p<0.01; ***p<0.001; NS-non significant

Apabetalone Reduces Monocyte Adhesion to Cytokine-Activated HBMVECs

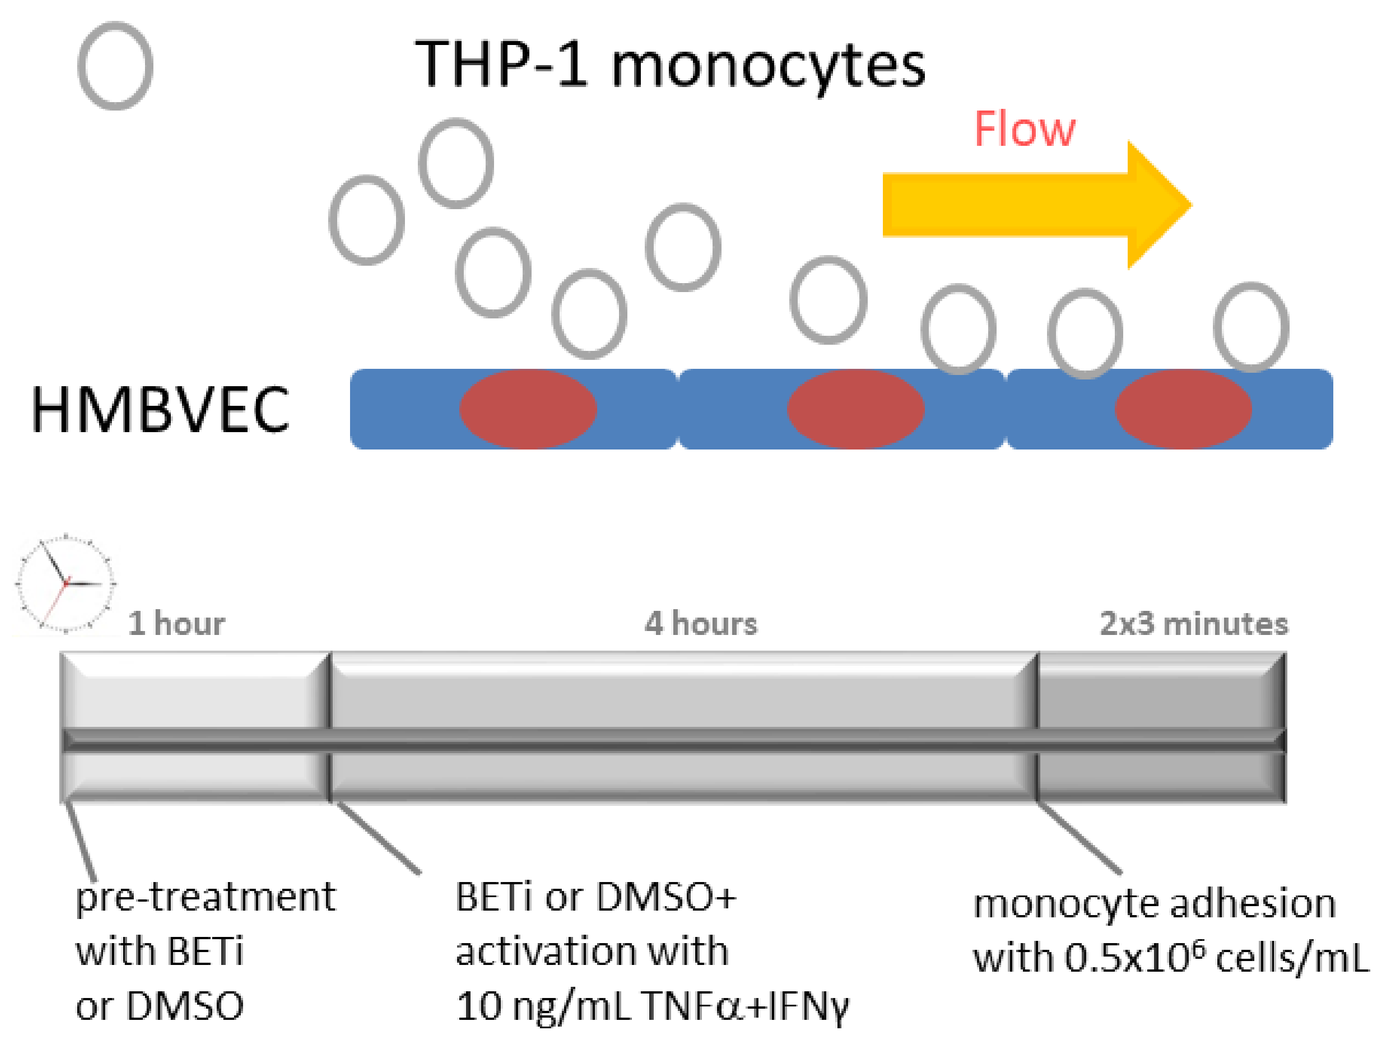
THP-1 monocytes
Flow
HMBVEC
1 hour
4 hours
2x3 minutes
pre-treatment with BETi or DMSO
BETi or DMSO+ activation with 10 ng/mL TNFα+IFNγ
monocyte adhesion with 0.5x10⁶ cells/mL

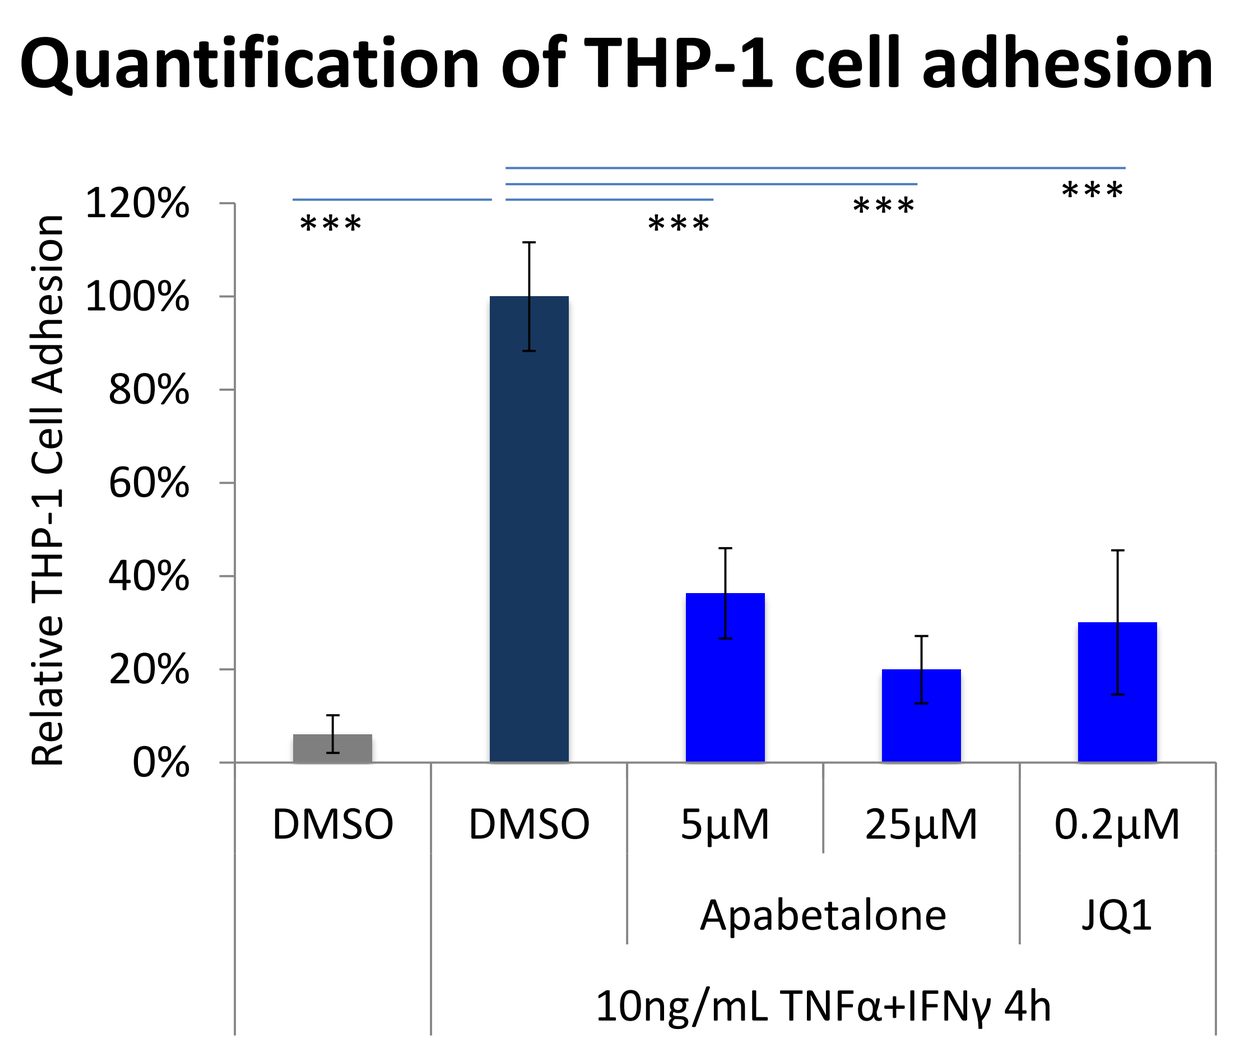
Quantification of THP-1 cell adhesion
Relative THP-1 Cell Adhesion
DMSO
5µM
25µM
0.2µM
Apabetalone
JQ1
10ng/mL TNFα+IFNγ 4h

Statistical significance determined with a Student's *t*-test * p<0.05; **p<0.01; ***p<0.001

Apabetalone Suppresses Expression of Inflammatory Mediators in BV-2 Microglial Cells

BV-2 cells were stimulated for 2h with 100 ng/mL LPS + 5 ng/mL IFNγ ± BETi. Gene expression was analyzed by real-time PCR.

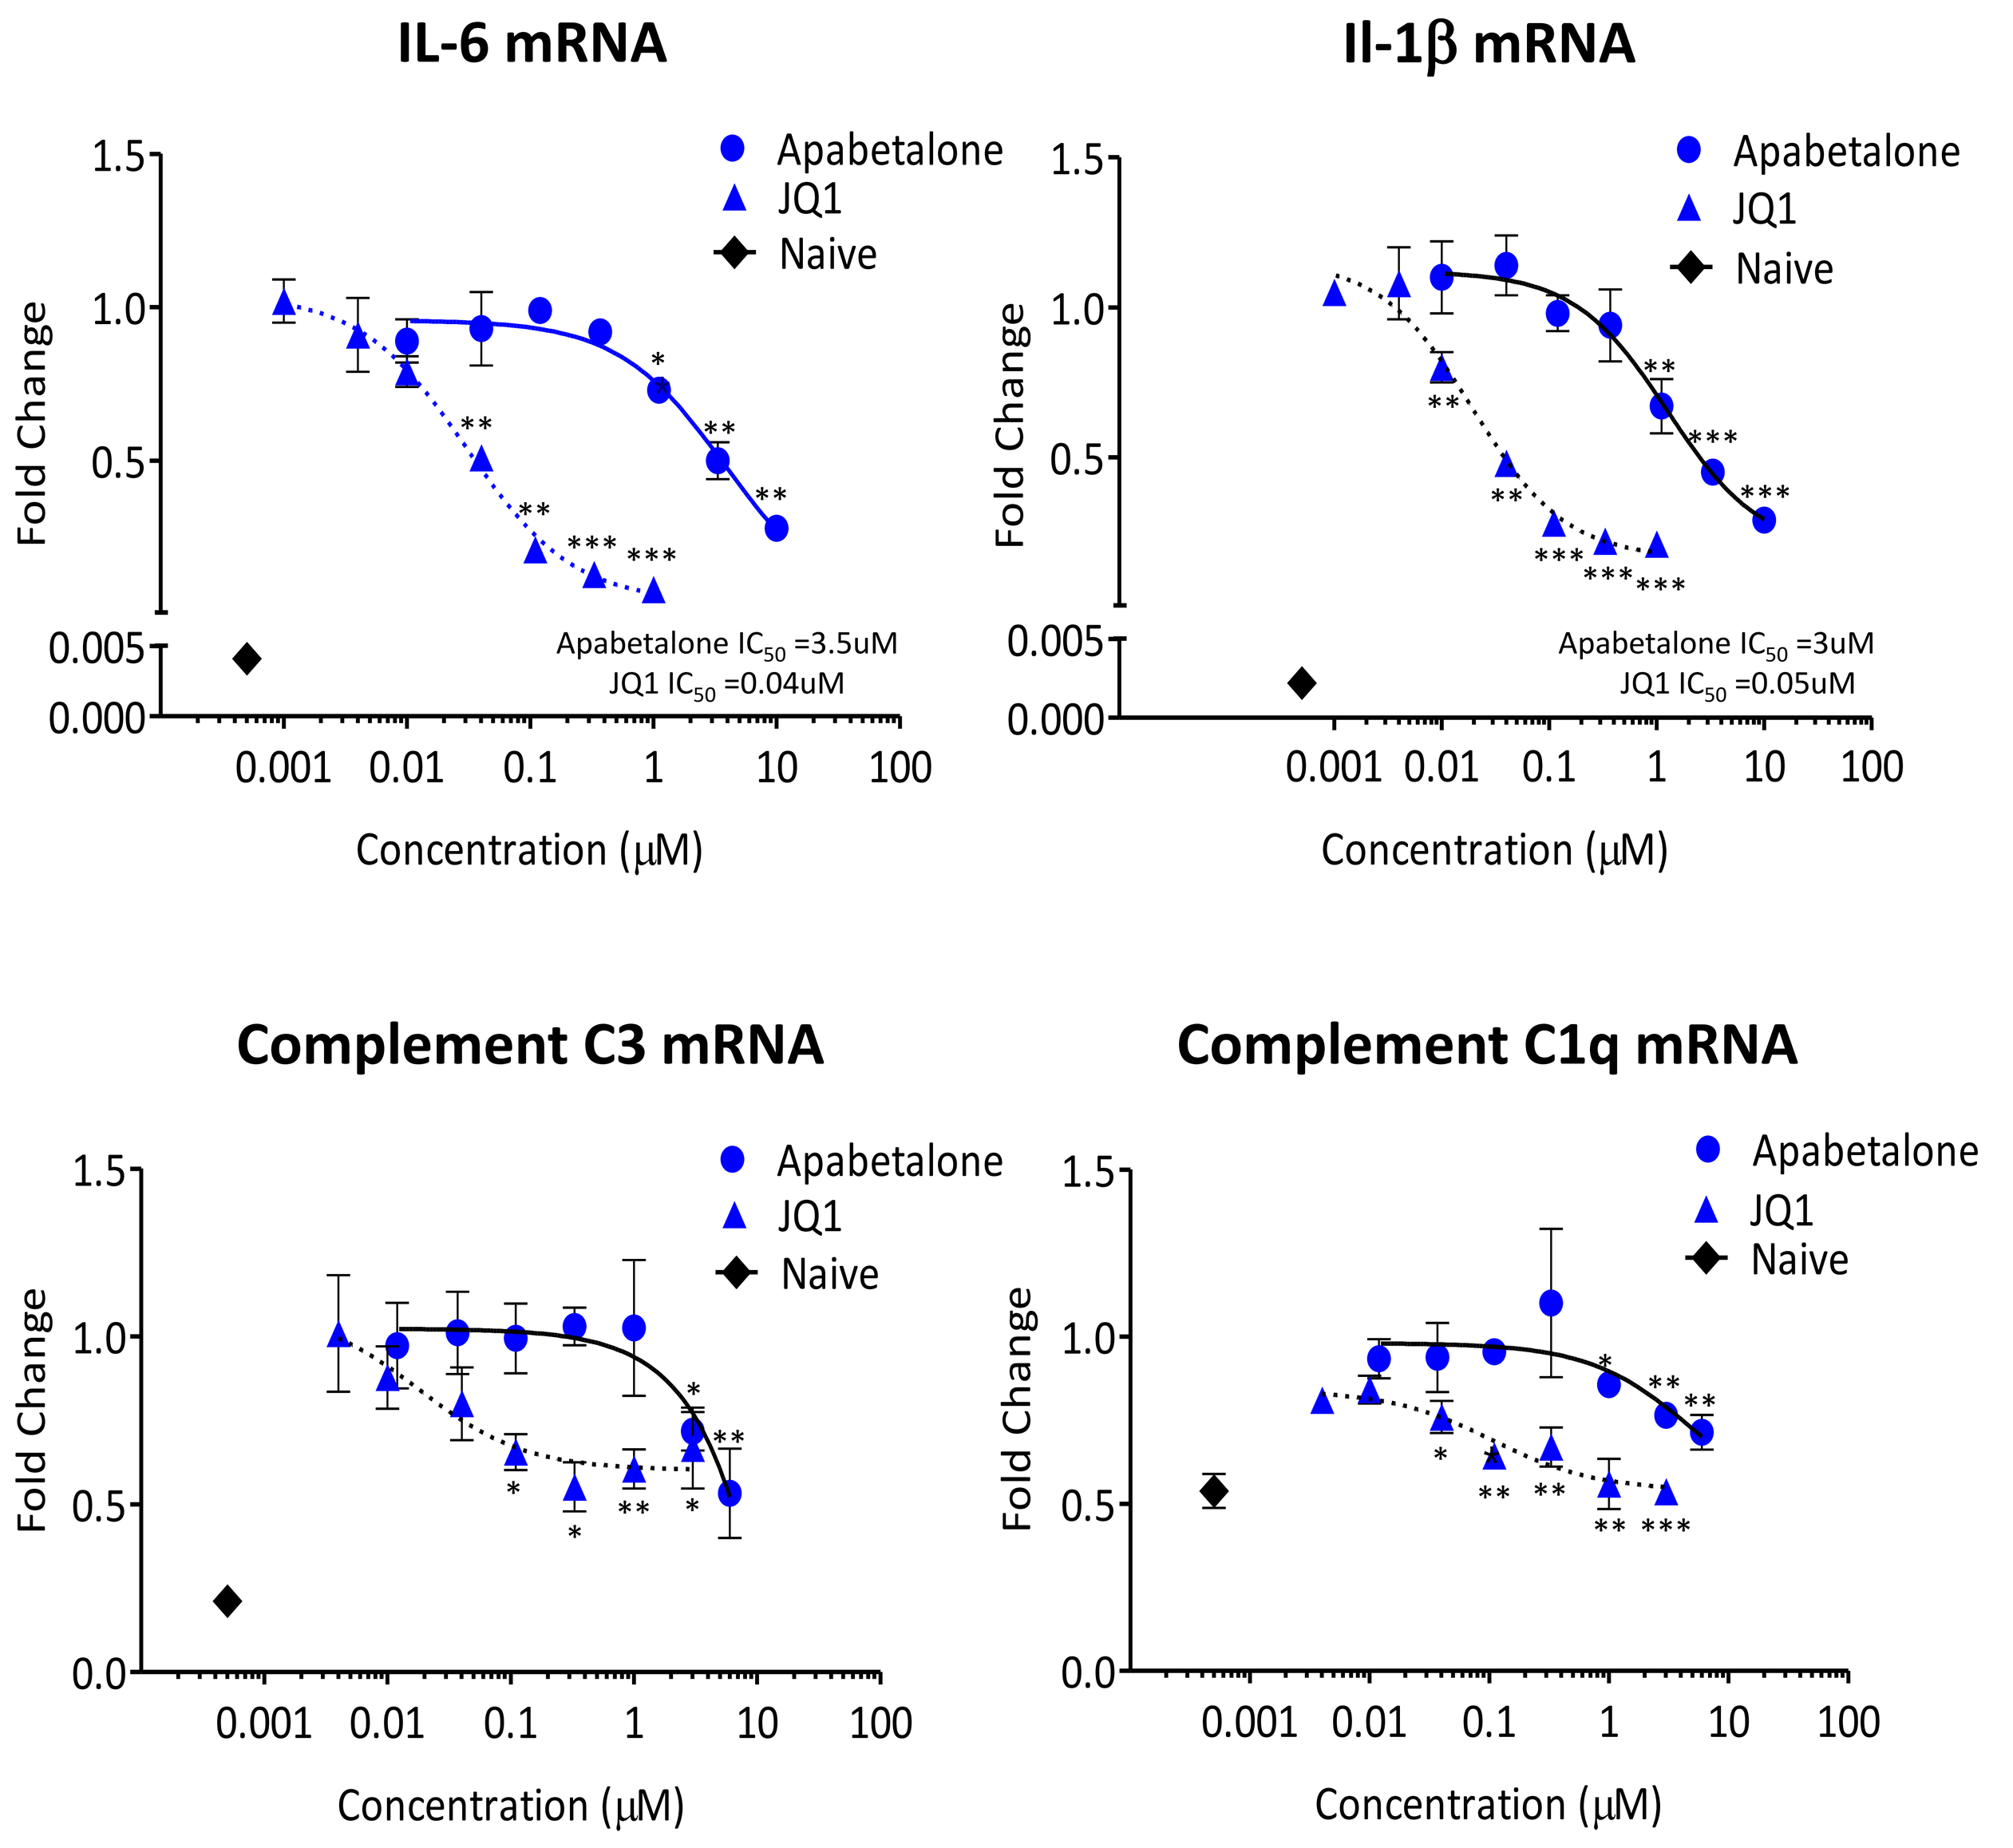
IL-6 mRNA
Il-1β mRNA
Complement C3 mRNA
Complement C1q mRNA
Apabetalone
JQ1
Naive
Fold Change
Concentration (µM)
Apabetalone IC50 =3.5uM
JQ1 IC50 =0.04uM
Apabetalone IC50 =3uM
JQ1 IC50 =0.05uM

Naïve cells did not receive cytokine or BETi treatment. Statistical significance determined with a Student's *t*-test * p<0.05; **p<0.01; ***p<0.001

Apabetalone Reduces Expression of Inflammation Markers in the Brain of Endotoxemic Mice

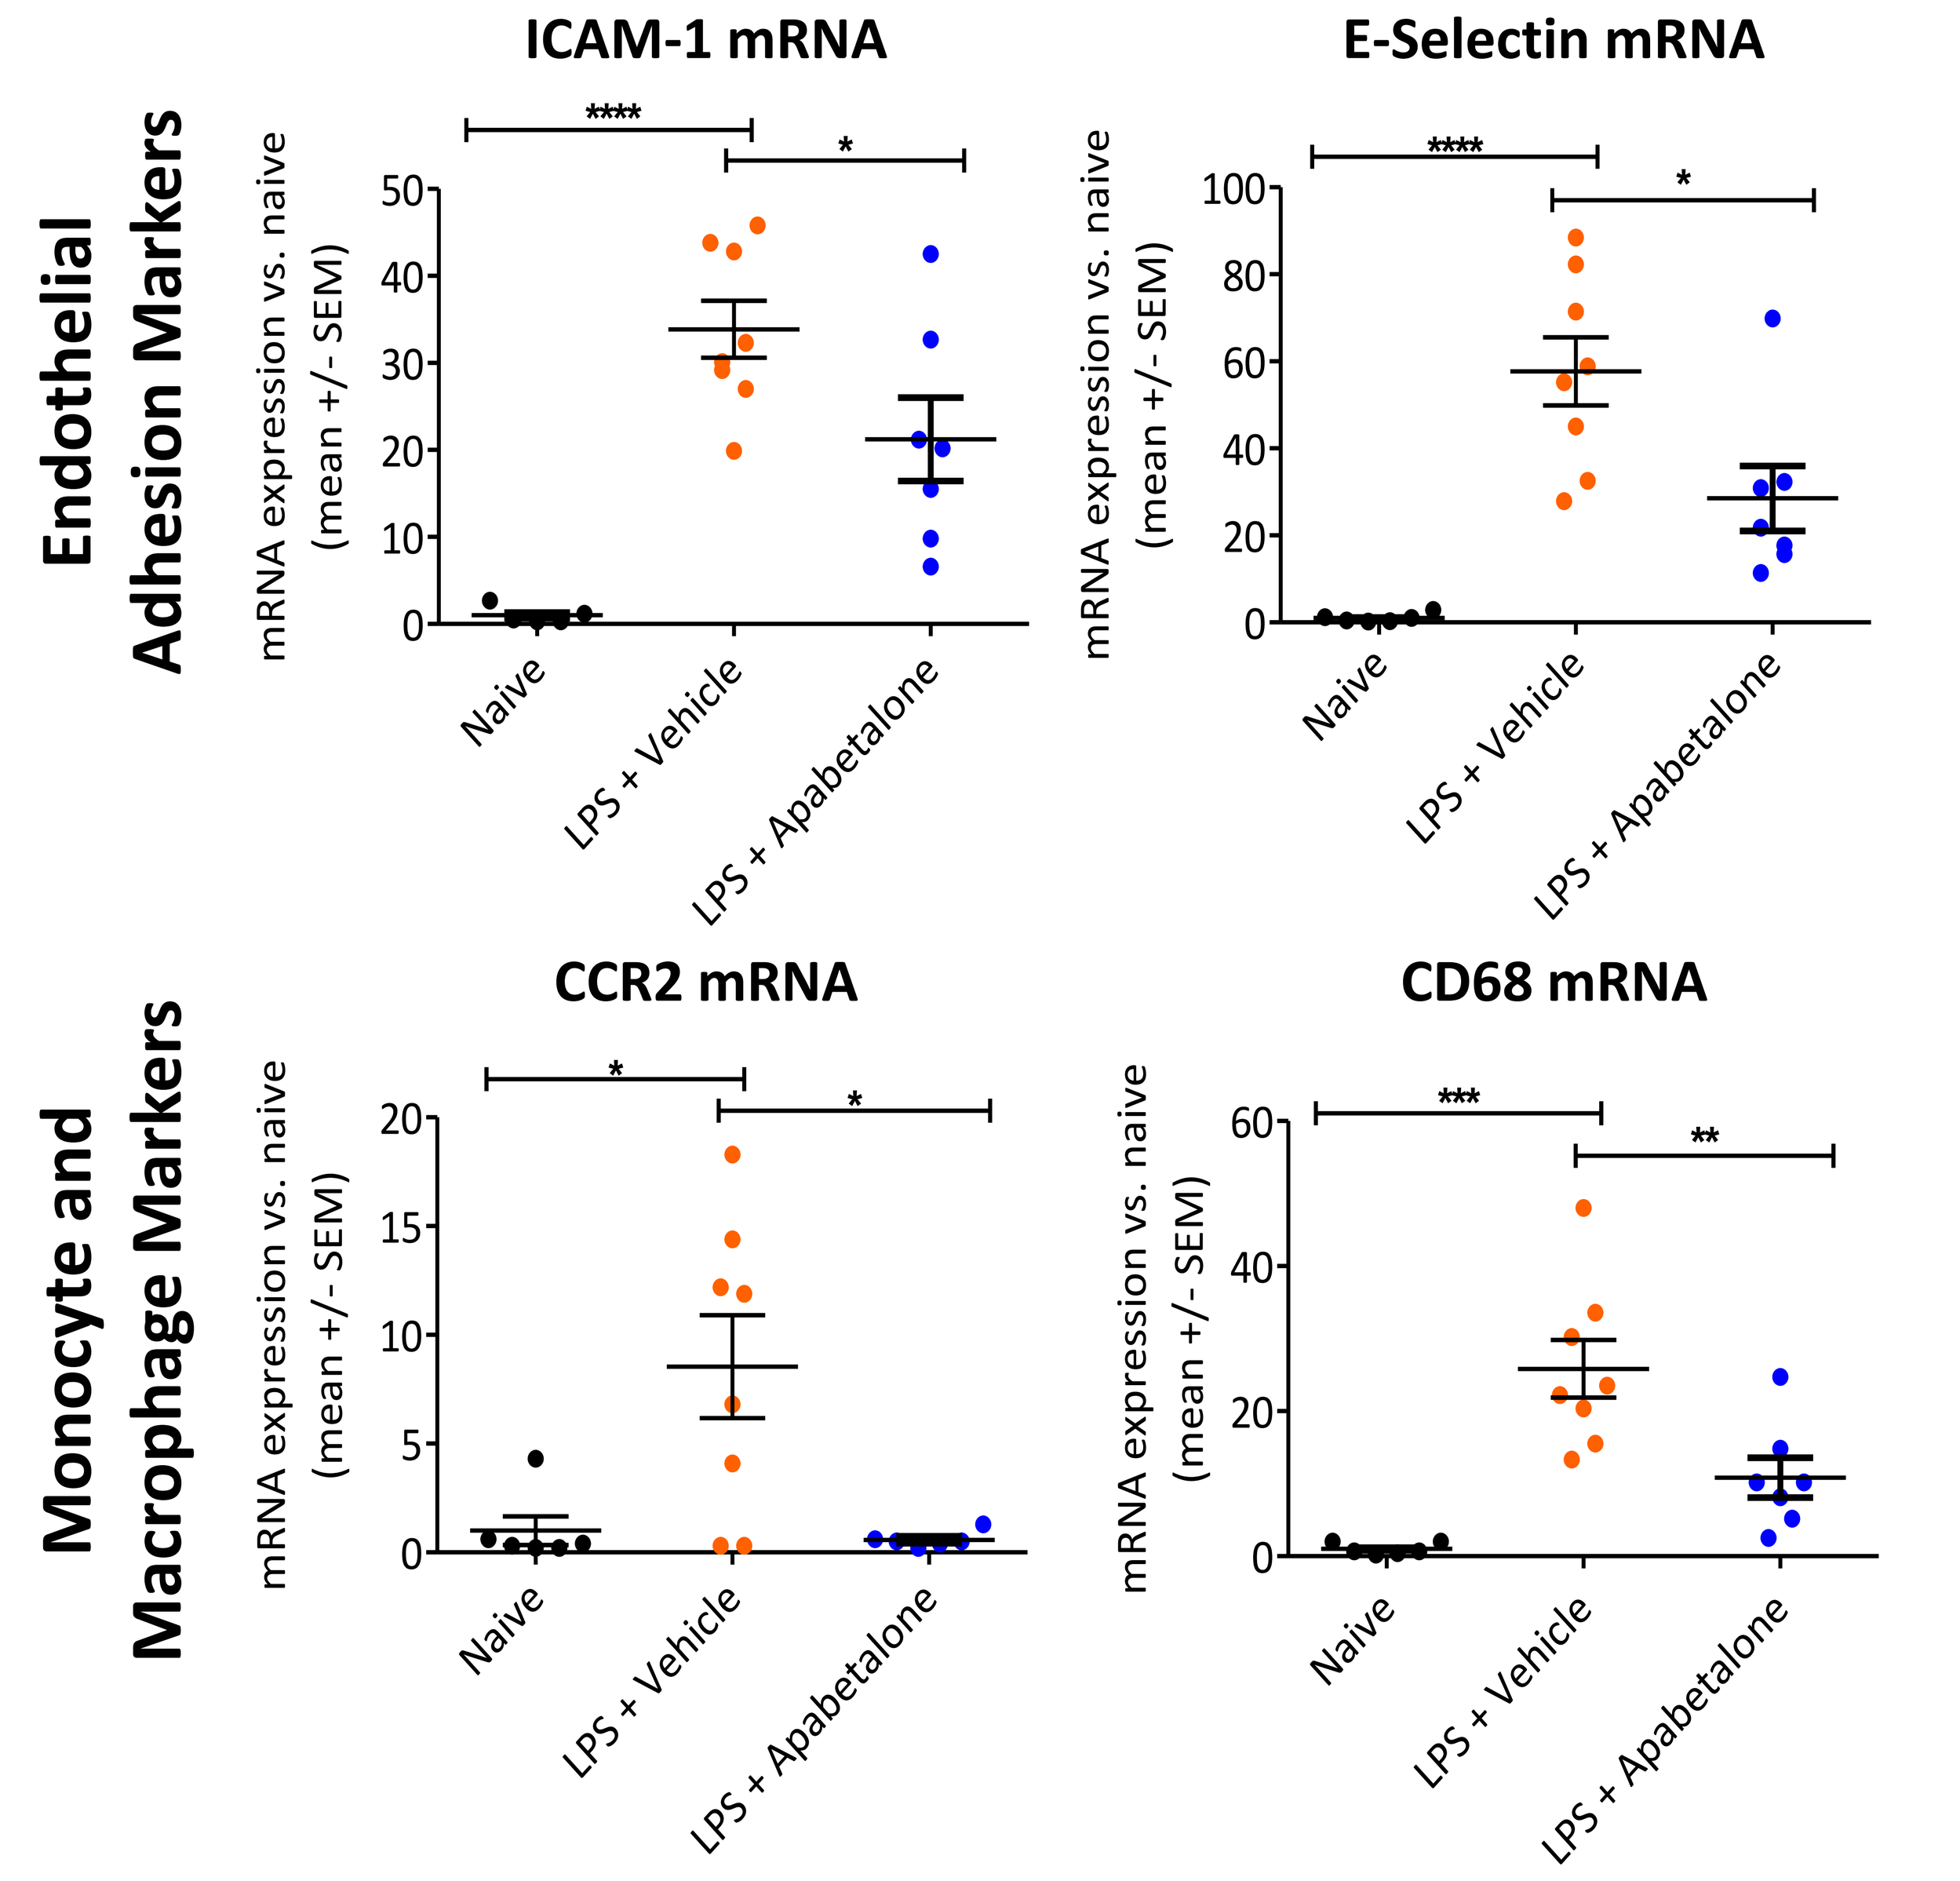
Endothelial Adhesion Markers
Monocyte and Macrophage Markers
ICAM-1 mRNA
E-Selectin mRNA
CCR2 mRNA
CD68 mRNA
mRNA expression vs. naive (mean +/- SEM)
Naive
LPS + Vehicle
LPS + Apabetalone

Naïve cells did not receive LPS or BETi treatment. Statistical significance determined with a Student's *t*-test * p<0.05; **p<0.01; ***p<0.001; ****p<0.0001

Conclusions

- In CNS cell models and endotoxemia mice, the epigenetic inhibitor apabetalone can counter inflammatory expression of cytokines, chemokines and markers of endothelial and microglial activation associated with neuroinflammation and cognitive dysfunction.
- The effect of apabetalone on cognition is currently being evaluated with MoCA in participants ≥70 years of age enrolled in the phase 3 BETonMACE trial focusing on cardiovascular outcomes in patients with CVD and diabetes.